From Crisis to Closure: Early Aggressive Debridement and Use of Skin Grafting in Limb Threatening Necrotizing Fasciitis

Hayley Collins, DPM, William Lopez, DPM, Michael Cooper, MD

Staten Island University Hospital; Staten Island, NY

Staten Island University Hospital
Northwell Health®

Purpose

This case highlights the potential for limb salvage through early aggressive surgical intervention and staged reconstruction, even in the context of profound systemic compromise and delayed presentation

Introduction

- Necrotizing fasciitis (NF) in the setting of diabetic ketoacidosis (DKA) and septic shock presents a formidable challenge, often necessitating major limb amputation. Early aggressive surgical debridement remains the cornerstone of NF management.
- Multiple studies underscore that time-to-operating room is a decisive factor in survival and limb preservation.
- Gelbard et al. (2018) found that delays beyond 12 hours from diagnosis significantly increased mortality and complication rates [2].
- Wong et al. (2024) described a 4 step approach to debridement—diagnosis, extent mapping, radical excision, and post-excision wound care—emphasizing that systematic, aggressive excision is essential to halt disease progression [3].
- In limb-threatening cases, especially involving the foot and ankle, early radical debridement can be limb-saving.
- Sevimli et al. (2016) reported that recurrent aggressive debridement, paired with interdisciplinary care, enabled limb preservation in a patient with extensive lower extremity NF [5].
- Advanced wound coverage techniques—including split-thickness skin grafting (STSG)—are increasingly viable following early source control.
- Zhu et al. (2019) documented successful STSG in a patient with Klebsiella pneumoniae NF and DKA, reinforcing that grafting is feasible even in metabolically unstable hosts [6].
- Early, aggressive debridement is not only life-saving but limb-saving, especially when paired with strategic reconstruction and interdisciplinary coordination.

Case Report

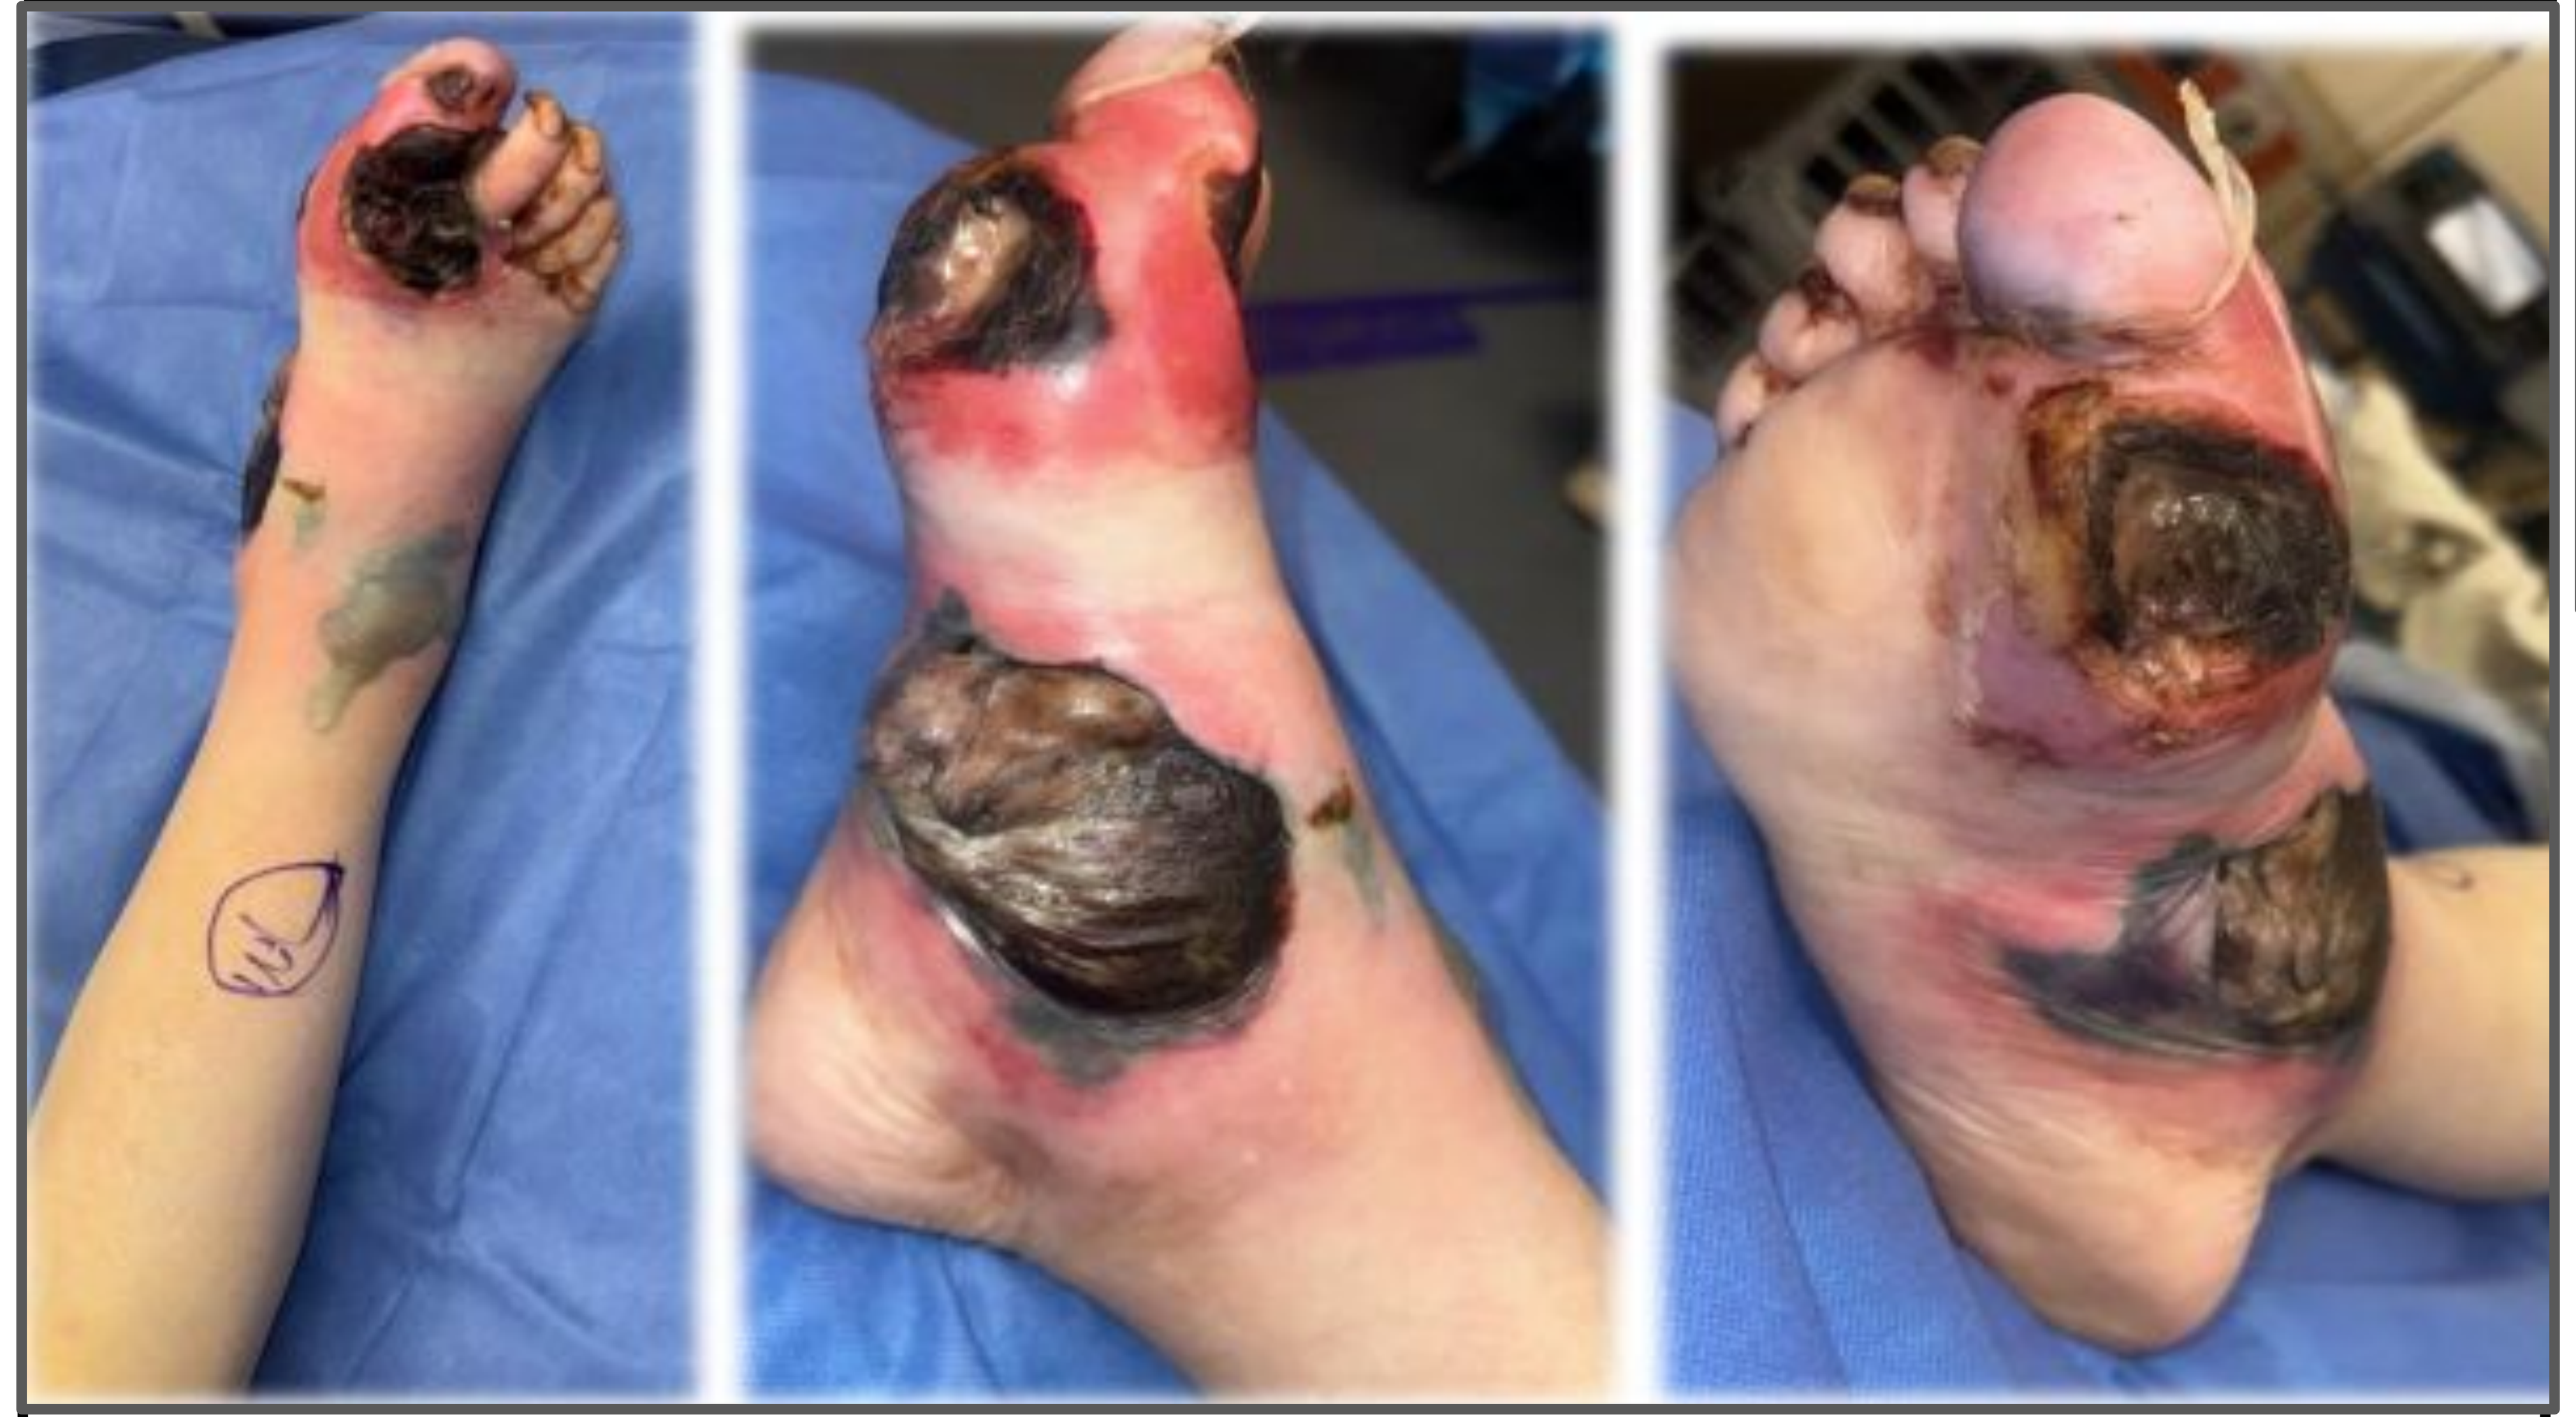

A 64-year-old female with a longstanding history of poorly controlled Type 2 Diabetes Mellitus-who had not engaged with medical care in over two decades—presented to the emergency department in diabetic ketoacidosis (DKA) and septic shock. She described a foul-smelling wound on her right lower extremity, which had worsened significantly in the past 24 hours. She endorsed localized pain in the affected limb but denied fevers or chills.

Domain	Findings	Clinical Implications
History	Fall, poor appetite, foul-smelling wound, delayed care, lives alone	High risk for delayed presentation and systemic compromise
Vitals	BP 73/67, HR 94, RR 18, Temp 98.1°F	Hemodynamic instability consistent with septic shock
Labs	WBC 38.47, Na^+ 119, Glucose 557	Severe leukocytosis, hyponatremia, and hyperglycemia in DKA
XR	Subcutaneous gas in medial foot and anterior tibial soft tissues	Suggestive of gas-forming infection; concern for NF
Duplex	No DVT or PAD	Confirms vascular access for limb salvage
CT- Bone	Osseous erosions, intraosseous gas in 1st metatarsal and phalanges	Osteomyelitis and septic arthritis
CT - ST	Extensive gas from ankle to proximal femur, large bullae, edema	Necrotizing fasciitis with deep fascial and muscular spread

Table 1. *Correlations of diagnostic findings and clinical implications concerning for NF.*

These findings were highly concerning for necrotizing fasciitis (Table 1). Sepsis protocol was initiated immediately, including aggressive fluid resuscitation, high-dose intravenous broad-spectrum antibiotics, and strict glucose control via continuous intravenous insulin infusion.

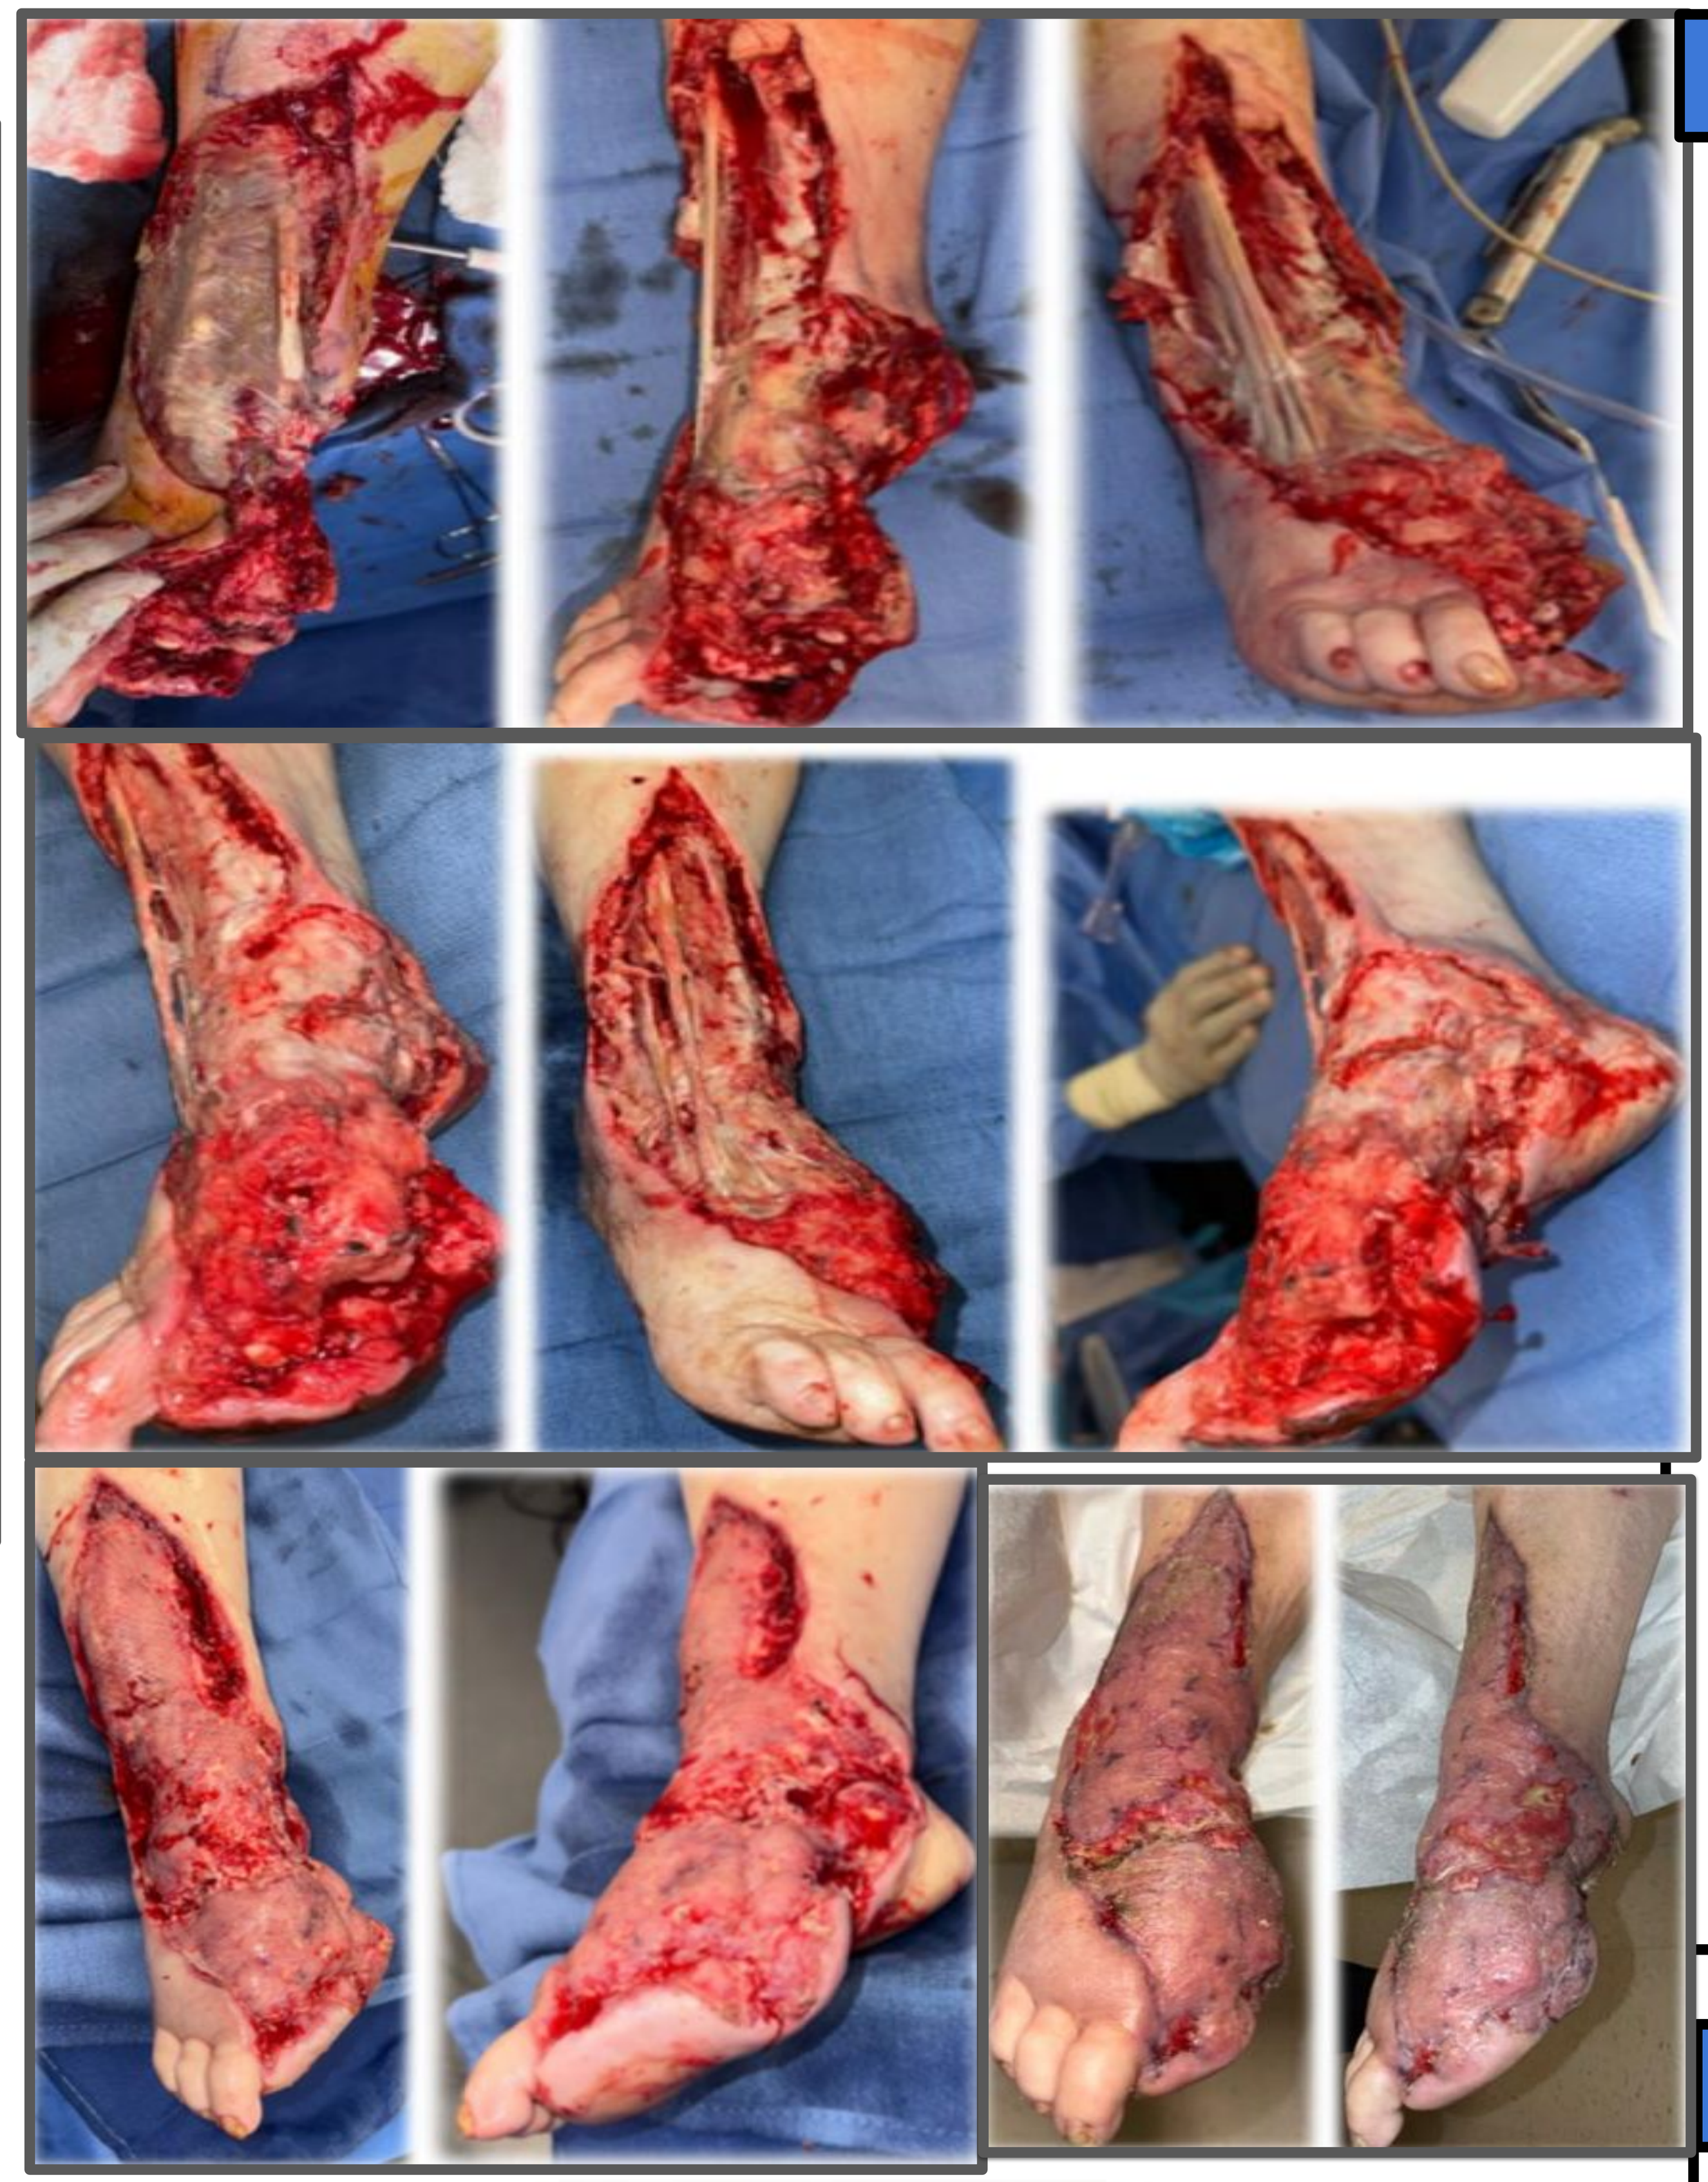

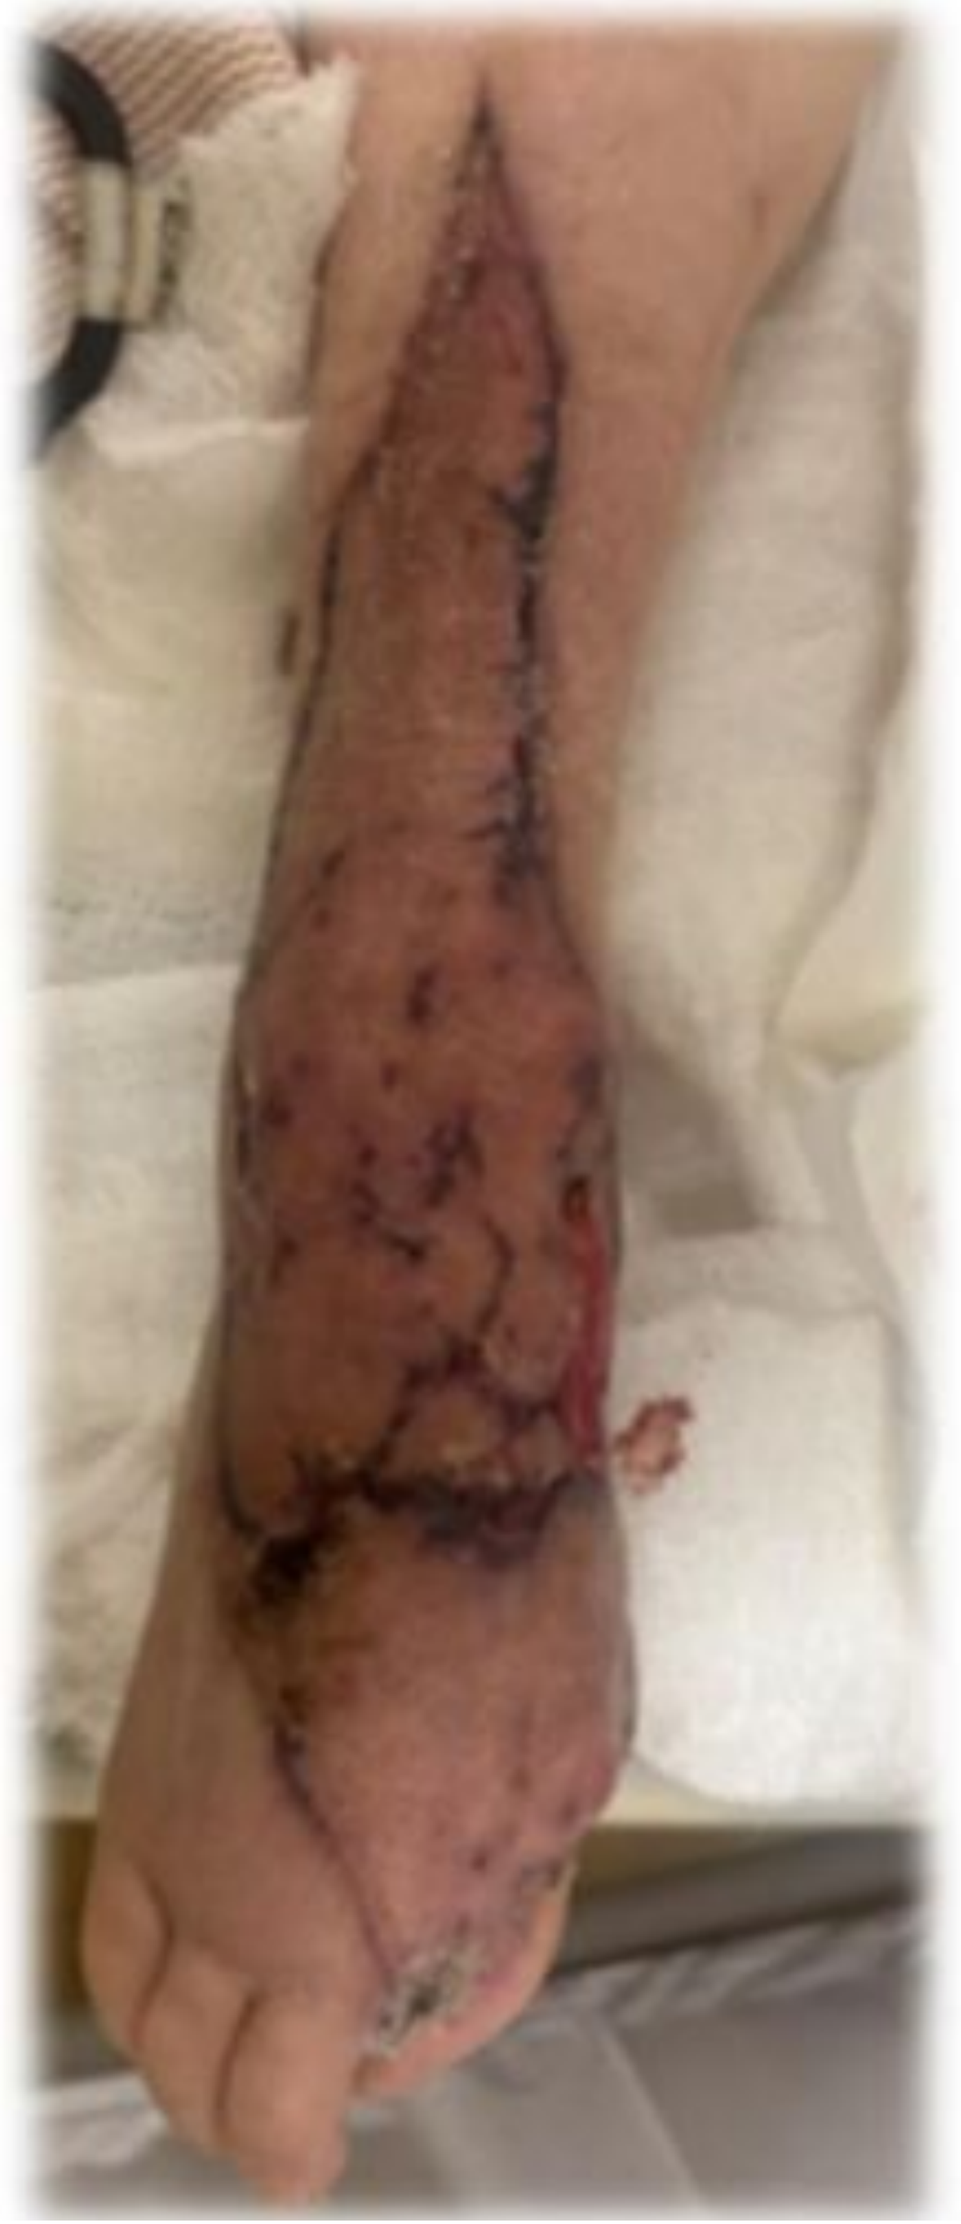

Results

1st Stage – Source Control:

- Emergent radical debridement → Involvement included the dorsal hallux, medial midfoot, and plantar first metatarsophalangeal joint, with hemorrhagic blistering and purulent drainage.
- The necrotic first ray and second digit were resected.
- Exposed midtarsal and subtalar joints with dishwater drainage and ankle joint purulence.
- A incision connected medial and lateral fasciotomy sites, allowing thorough debridement of necrotic fascia, muscle, and skin using electrocautery and a hydrosurgical debridement system.
- Cultures/Biopsies: E.Coli, E.Faecalis, and MRSA.

2nd Stage – Further Debridement & STSG:

- Returned to the operating room 72 hours later, where devitalized tissue of the RLE were debrided down to tendon
- By the third debridement (2 weeks later) robust granulation tissue had developed across previously exposed structures.
- STSG was harvested from the right thigh, meshed 1.5:1 and stapled down.

Conclusion

This case illustrates that even in profound compromise, early aggressive intervention can shift the trajectory from crisis to recovery. Radical debridement within hours prioritized source control and fascial decompression, aligning with literature emphasizing time-to-OR as critical for survival and limb preservation.

Despite initial concern for non-salvageability, dynamic surgical planning and interdisciplinary collaboration enabled limb salvage through four serial debridements. STSG with negative pressure therapy achieved >90% graft take by POD 14, supporting its use in metabolically unstable hosts when paired with meticulous wound prep and team-based care.

References

. Cheng NC, et al. Necrotizing fasciitis in patients with diabetes mellitus: clinical characteristics and risk factors for mortality. BMC Infect Dis. 2015;15:417.
2. Gelbard RB, et al. Timing of Operative Debridement for Necrotizing Soft Tissue Infections. J Trauma Acute Care Surg. 2018;85(1):208–214. Link
3. Wong CH, et al. Approach to Debridement in Necrotizing Fasciitis. Singapore General Hospital. 2024. PDF
4. Narayanan AS, et al. Surgical Strategies: Necrotizing Fasciitis of the Foot and Ankle Treated With Dermal Regeneration Matrix for Limb Salvage. Foot & Ankle Int. 2021;42(1):107–114. Link
5. Sevimli R, et al. Limb Salvaging Approach to Necrotizing Fasciitis: Recurrent Aggressive Debridement. Ann Case Rep. 2016. Link
6. Zhu W, et al. Monomicrobial Klebsiella pneumoniae Necrotizing Fasciitis with Diabetic Ketoacidosis: A Case Report and Review of Literature. Int J Clin Exp Med. 2019;12(8):10962–10967

Ulcer Unplugged: How Detour Bypass and Oxygen Therapy Accelerate Repair

Hayley Collins DPM, William Lopez DPM, Mitchell Weinberg MD
Staten Island University Hospital; Staten Island NY

Purpose

To demonstrate the synergistic impact of combining topical oxygen therapy (TOT) with percutaneous transvenous femoropopliteal bypass in the longitudinal management of complex diabetic foot ulcers and advanced peripheral arterial disease.

Background

Topical oxygen therapy (TOT) is an emerging adjunct in the treatment of chronic wounds, particularly those complicated by vascular insufficiency. Oxygen supports key phases of healing—angiogenesis, collagen synthesis, epithelialization, and immune defense. A 2024 meta-analysis demonstrated that TOT significantly improves healing rates compared to standard care, with relative risks of 1.77 in randomized trials and 2.15 in observational studies. Advances in delivery systems now allow for continuous or cyclical oxygen application, enhancing outcomes in hypoxic wound beds. International consensus guidelines endorse TOT as part of multidisciplinary wound care, emphasizing appropriate patient selection.

Percutaneous transvenous femoropopliteal bypass offers a novel endovascular solution for long-segment superficial femoral artery (SFA) disease, particularly in cases of chronic total occlusion or in-stent restenosis. Unlike traditional open bypass, this technique uses a minimally invasive route to achieve durable revascularization. Pooled data from multicenter trials (n=273) demonstrated high technical success, low adverse event rates, and sustained patency. Final 3 year results reported primary patency of 58.2%, freedom from target lesion revascularization at 66.8%, and a major amputation rate of just 1.5%.

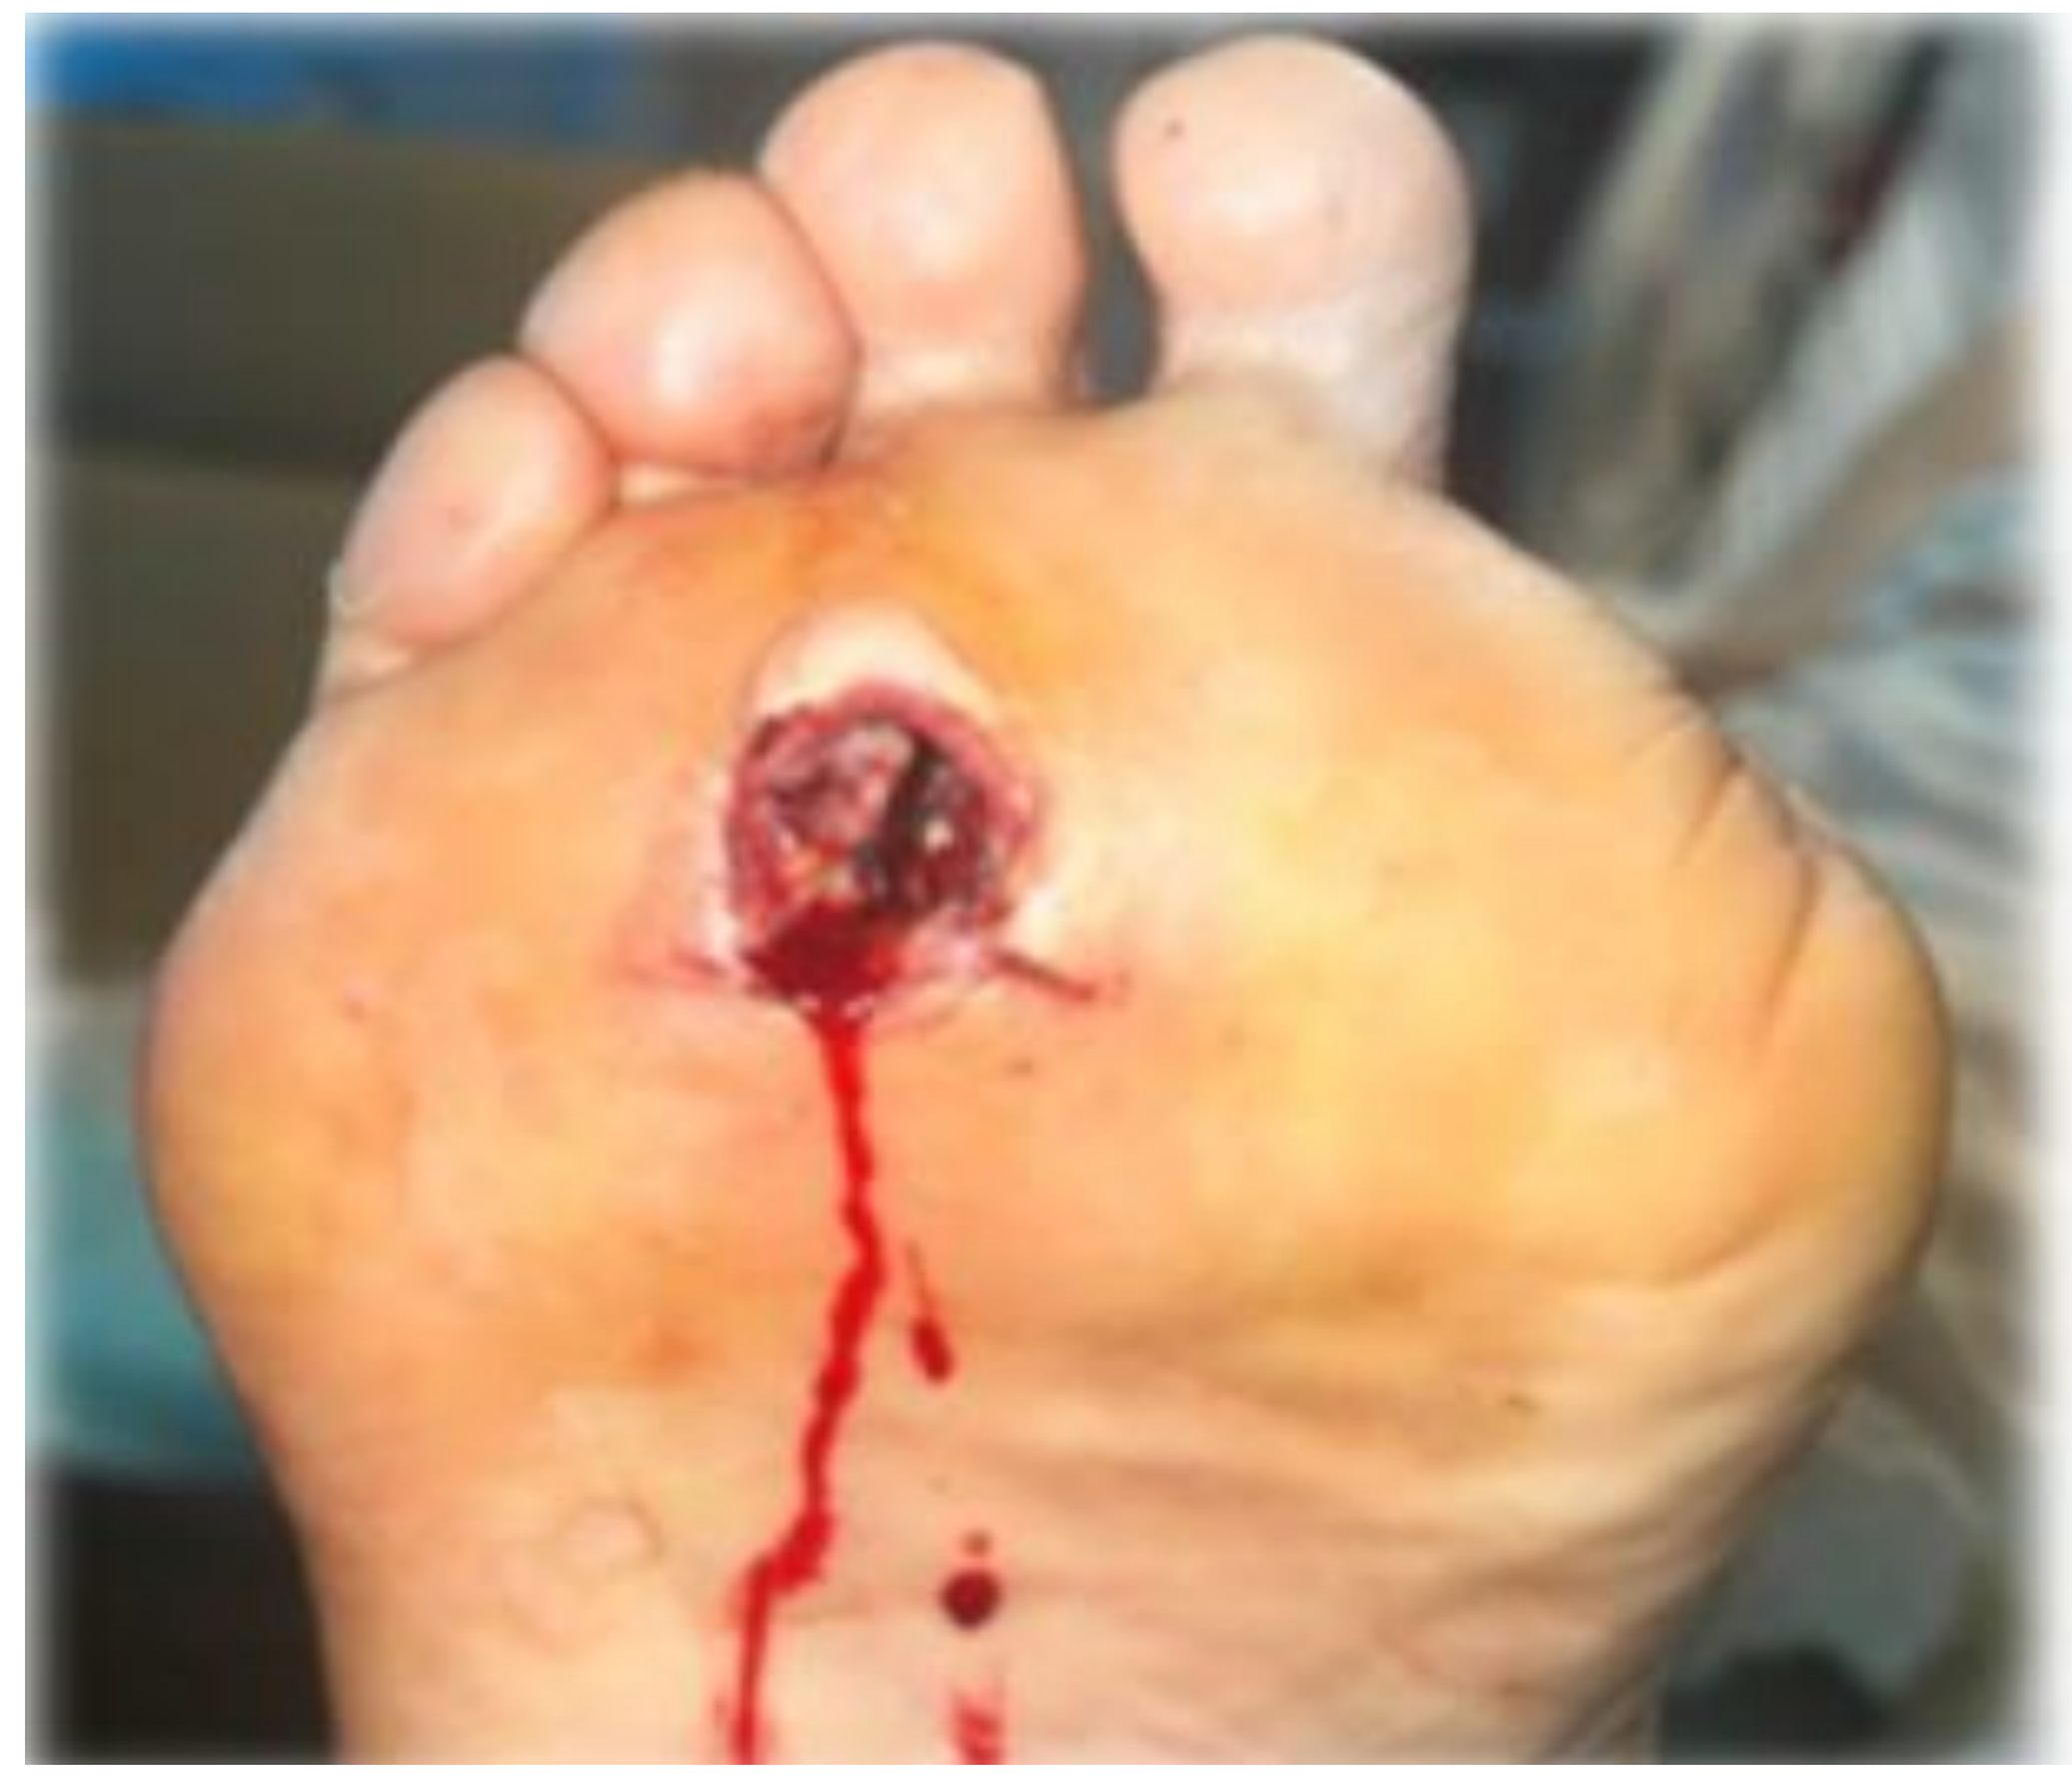

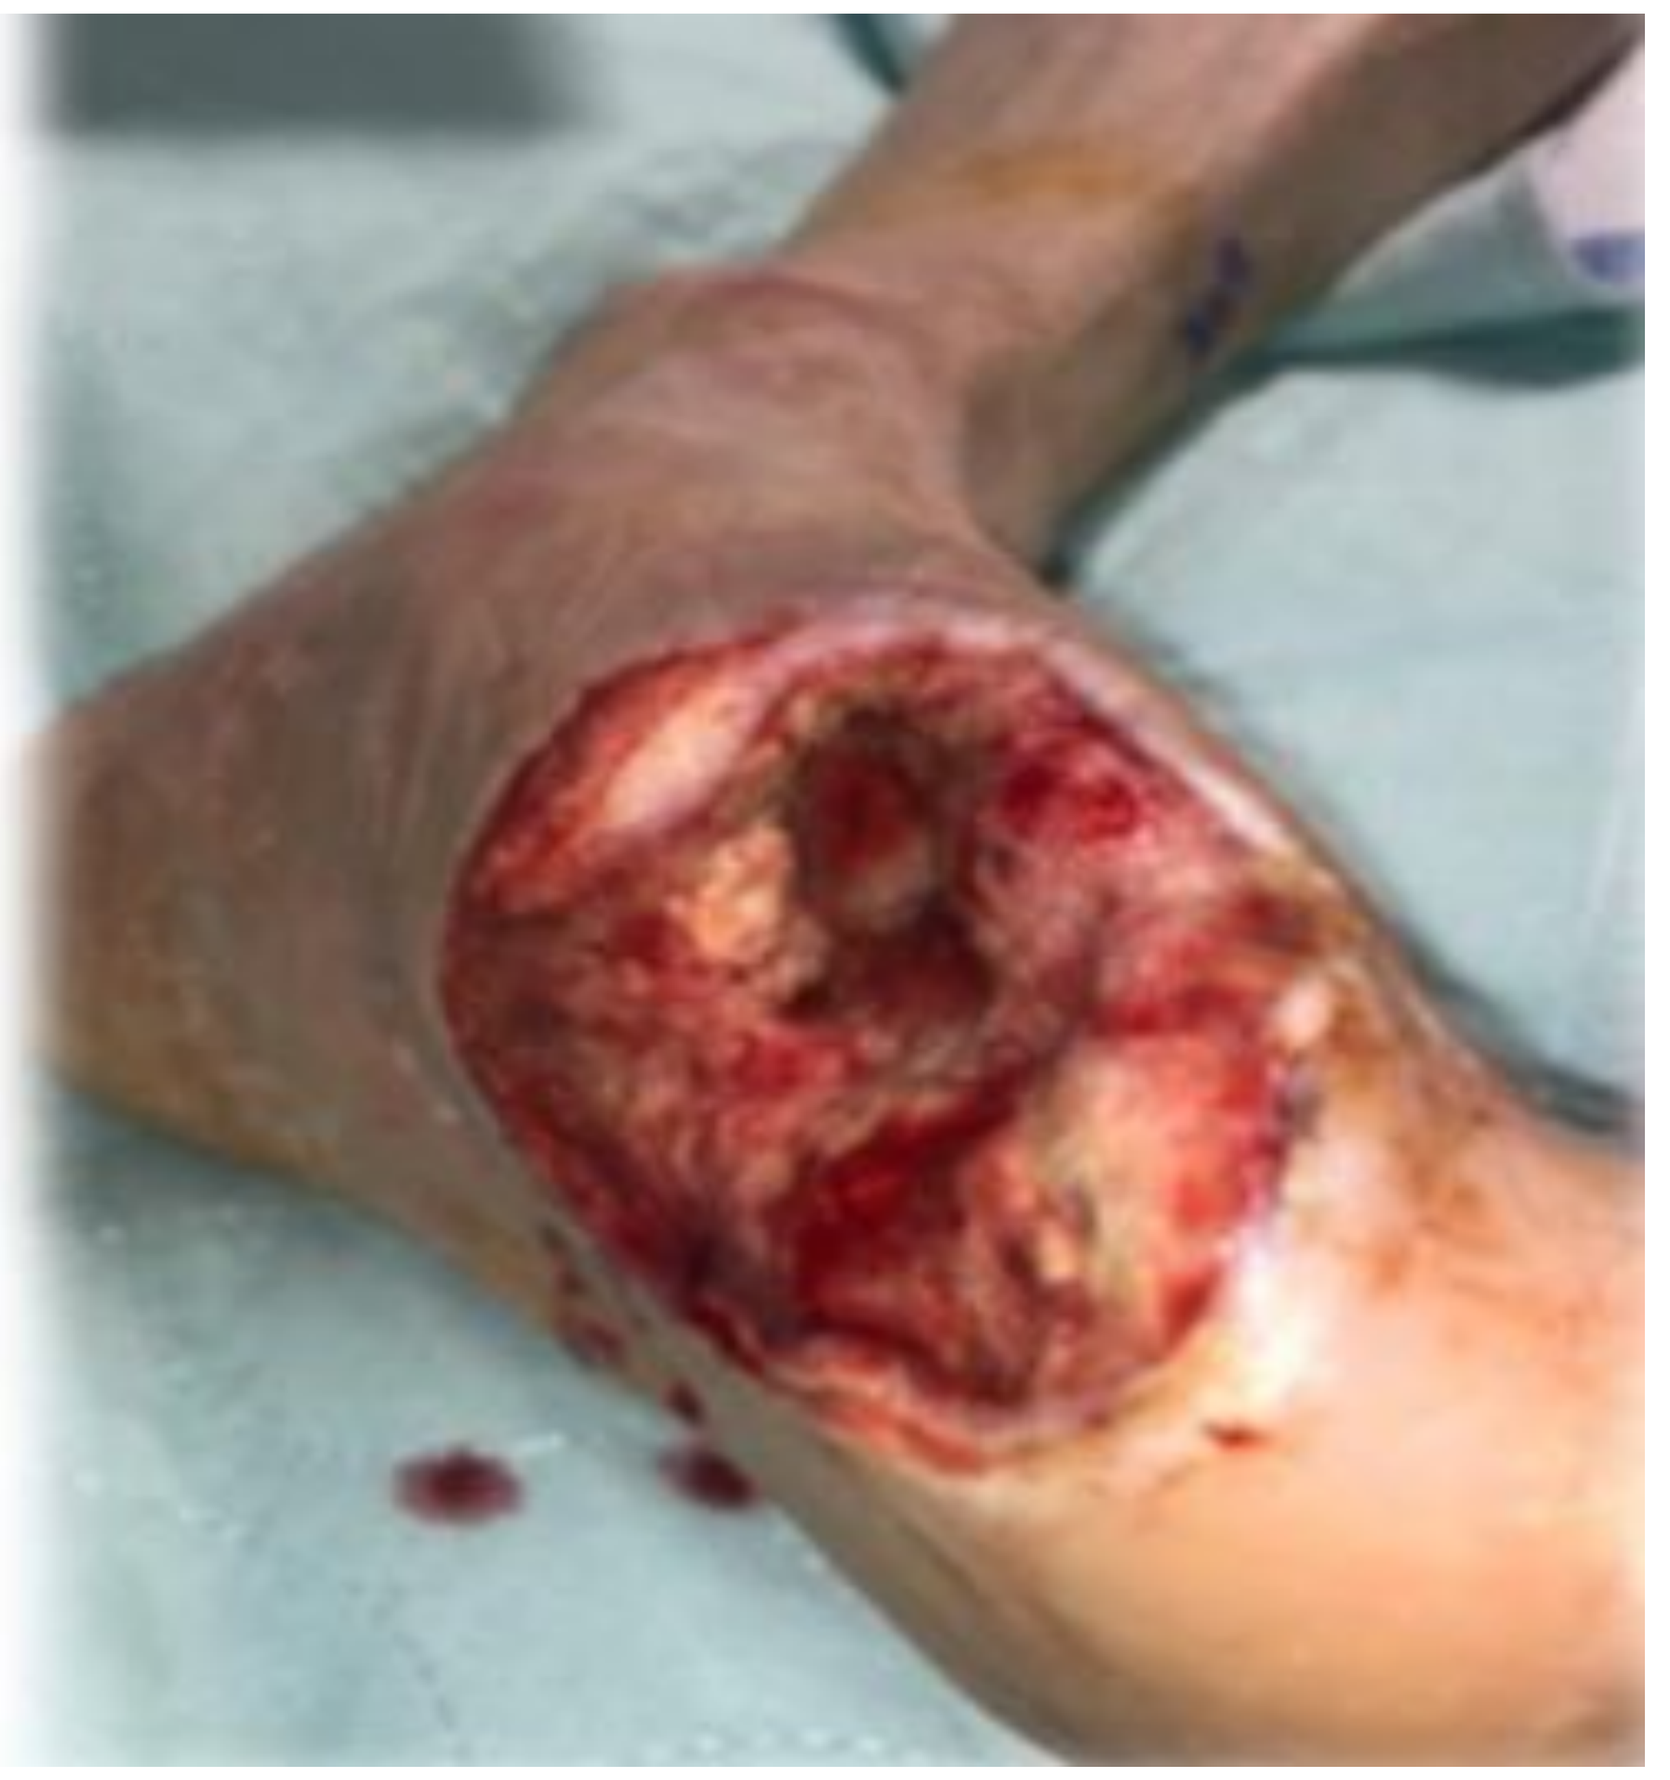

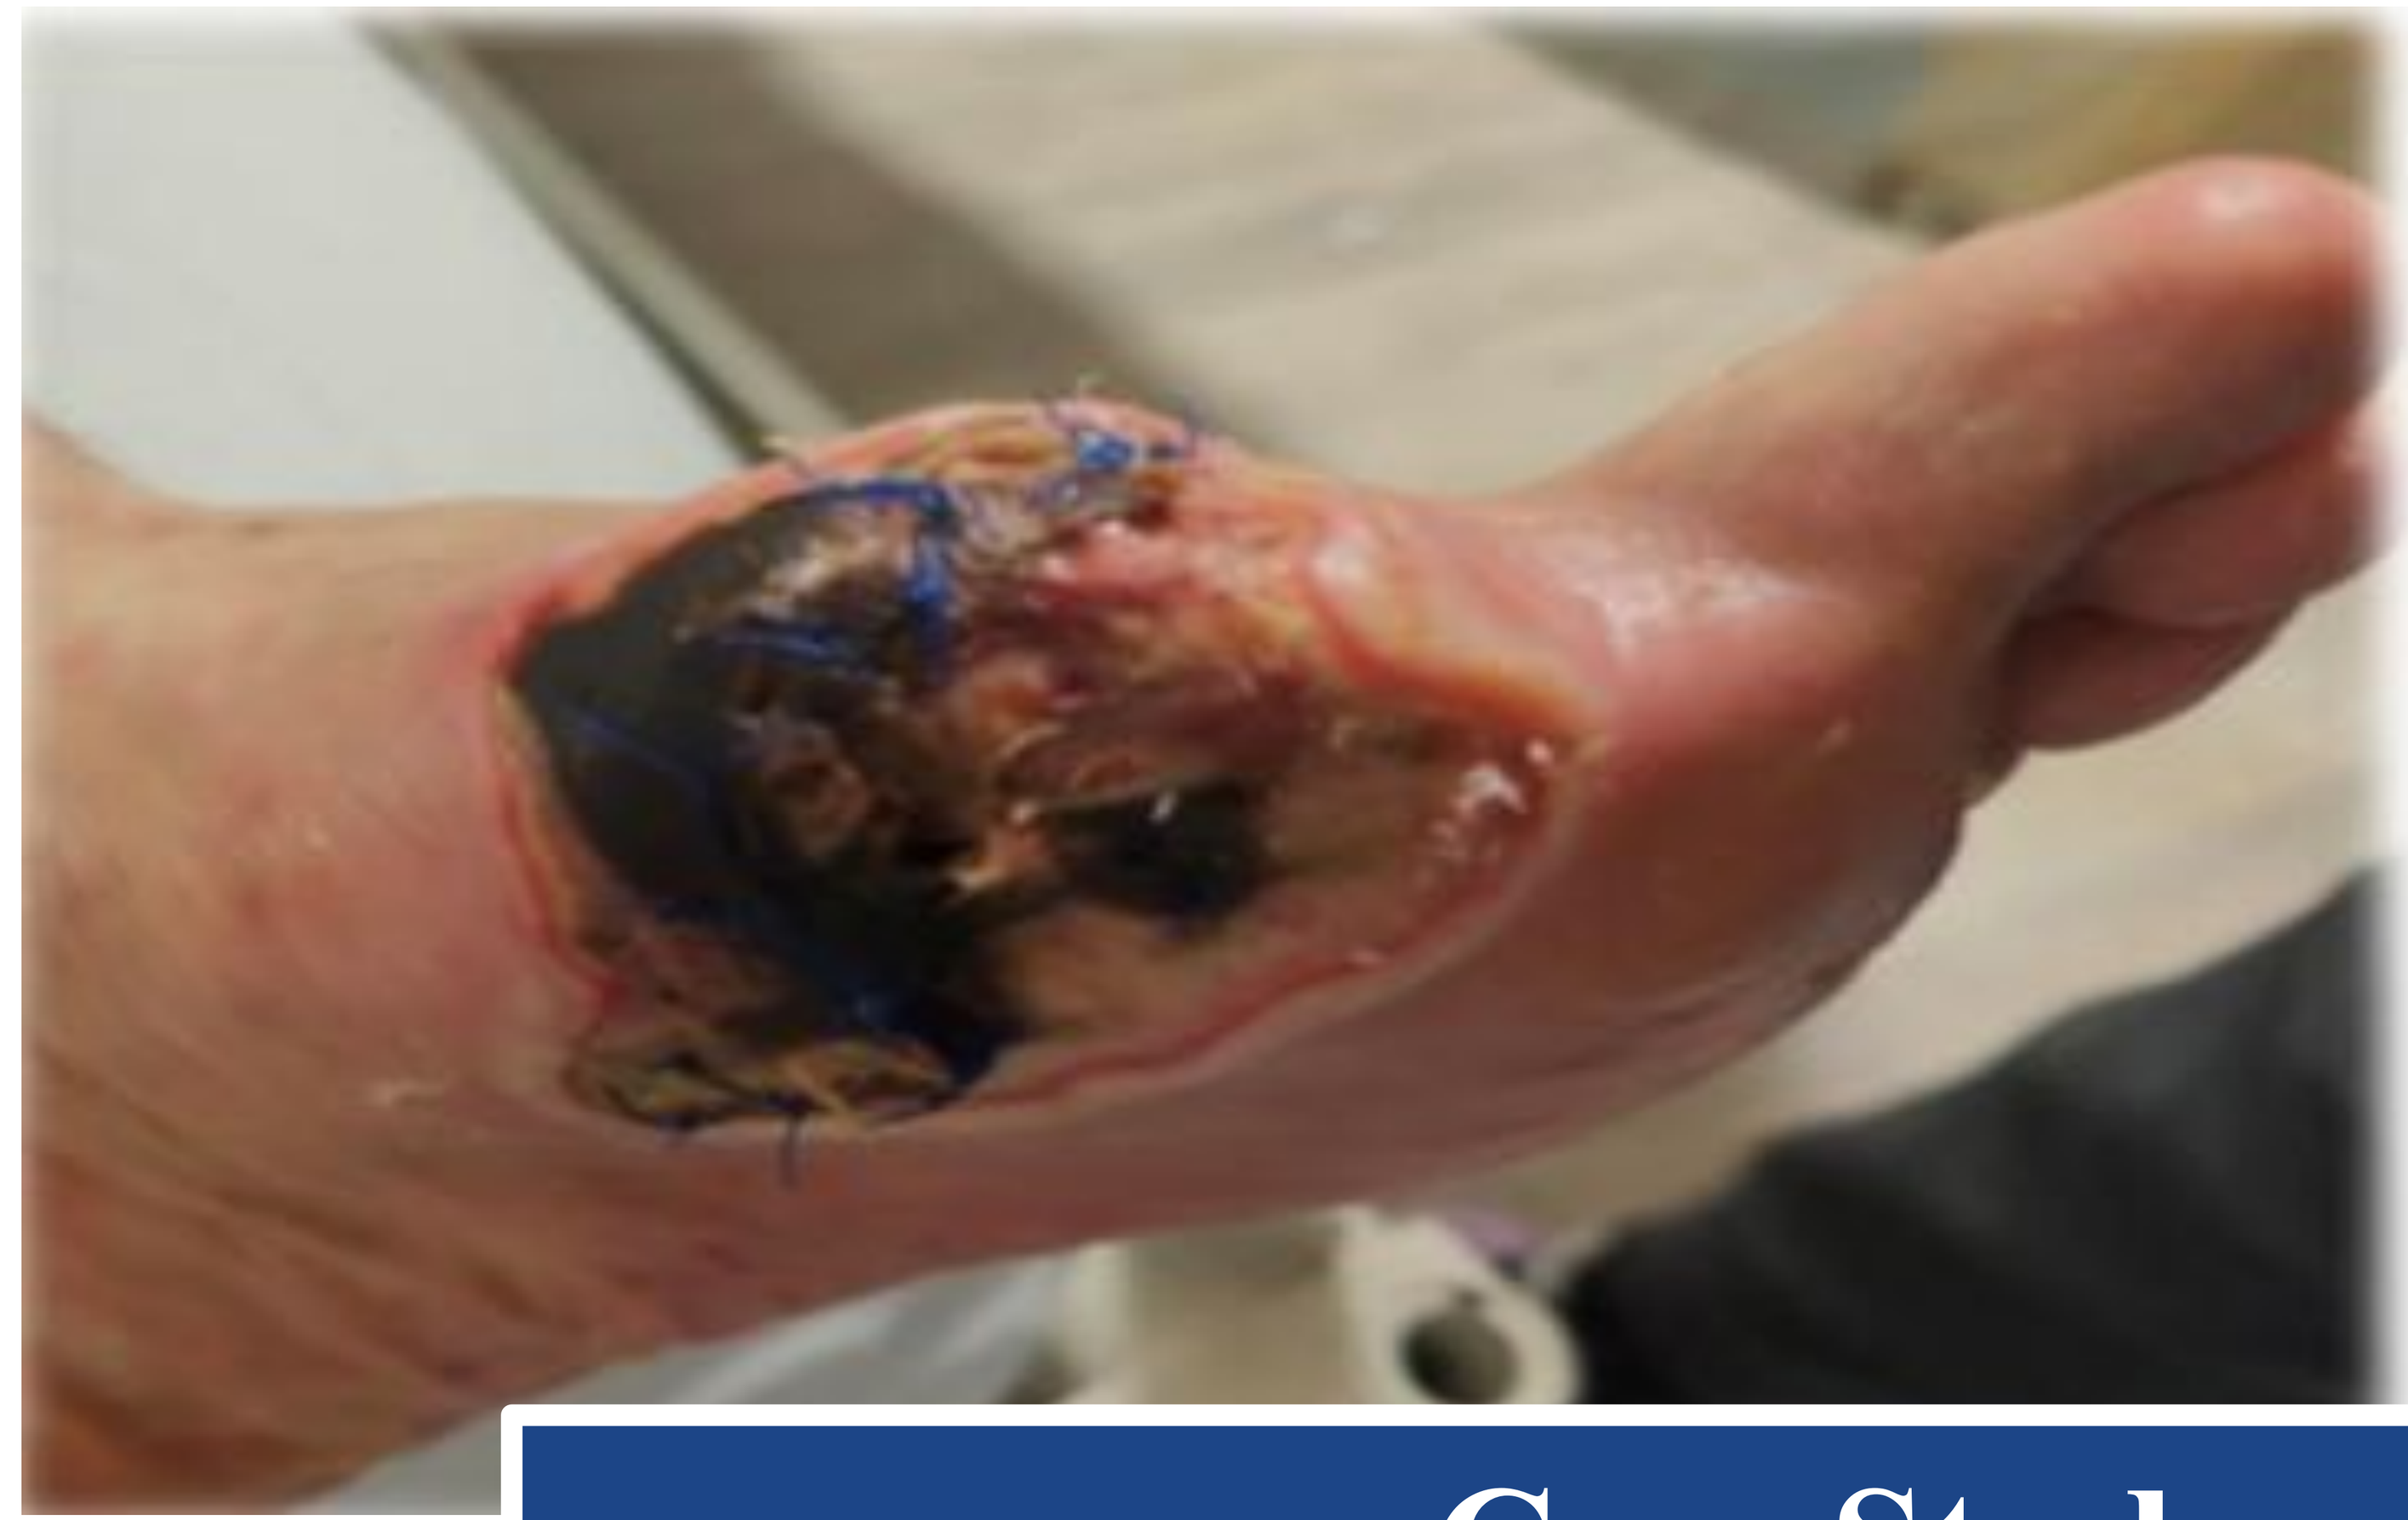

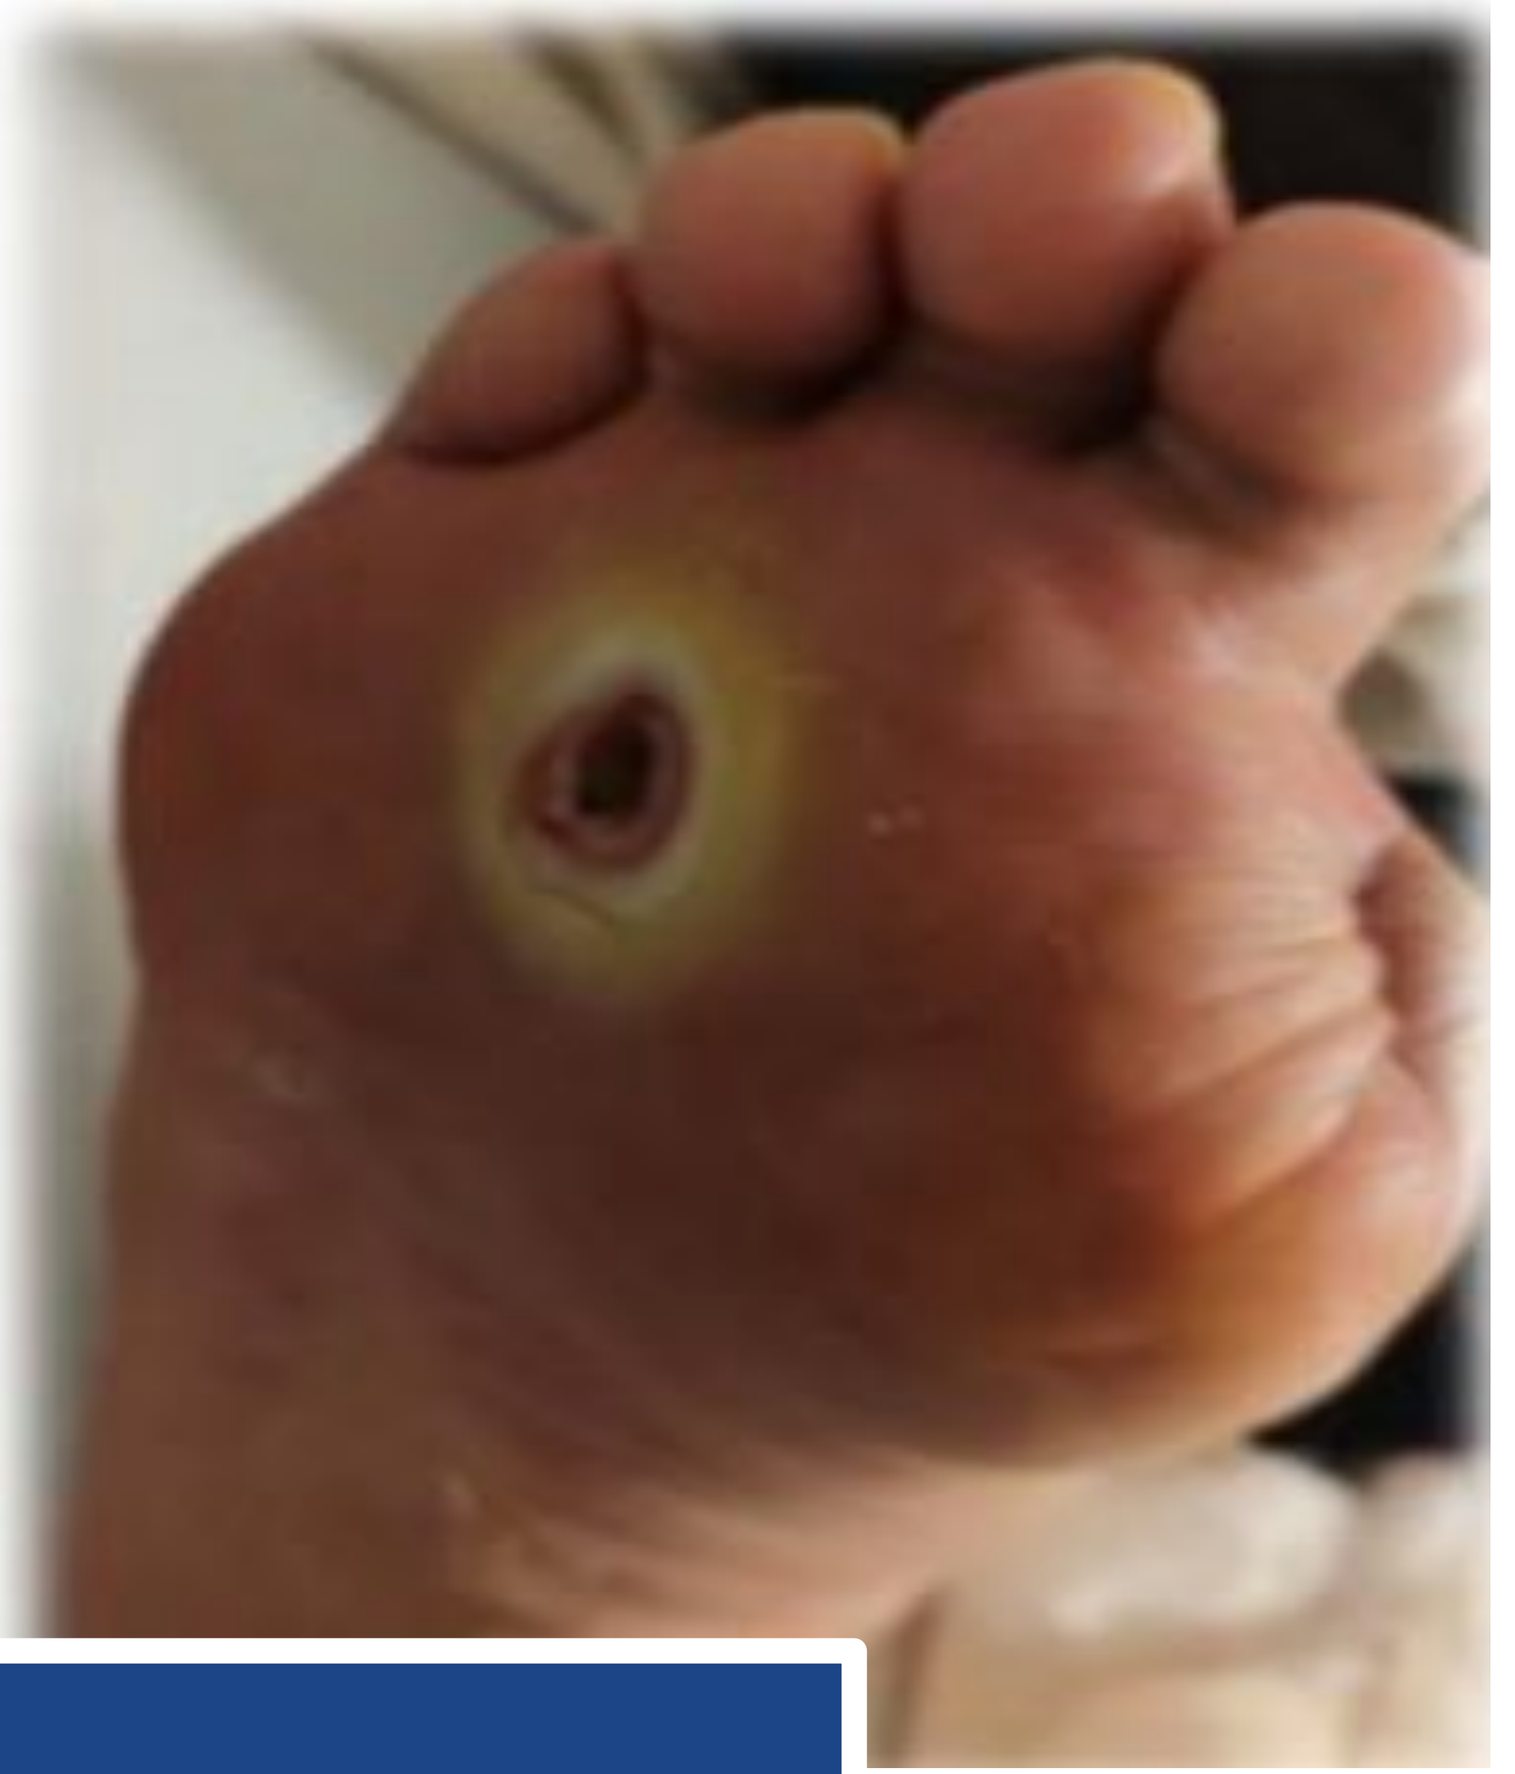

Case Study

A 68-year-old male with HTN, DM, HLD, and CAD has been followed for progressive PAD and recurrent ulcerations. Over the past year, he developed non-healing wounds and worsening claudication. Wound dehiscence was noted at the left hallux amputation site, with avascular necrosis and fibrotic base. A second ulcer under the right third metatarsal head showed purulent drainage and a sinus tract with probe-to-bone.

His vascular history included prior left-sided revascularization of the SFA, popliteal, and anterior tibial arteries, and PTA with three coronary DES to the right anterior tibial artery. Additional interventions included laser atherectomy and DCB to the left SFA and profunda. In 2024, he developed 90% stenosis of the right SFA and ISR of the left SFA/POP segment.

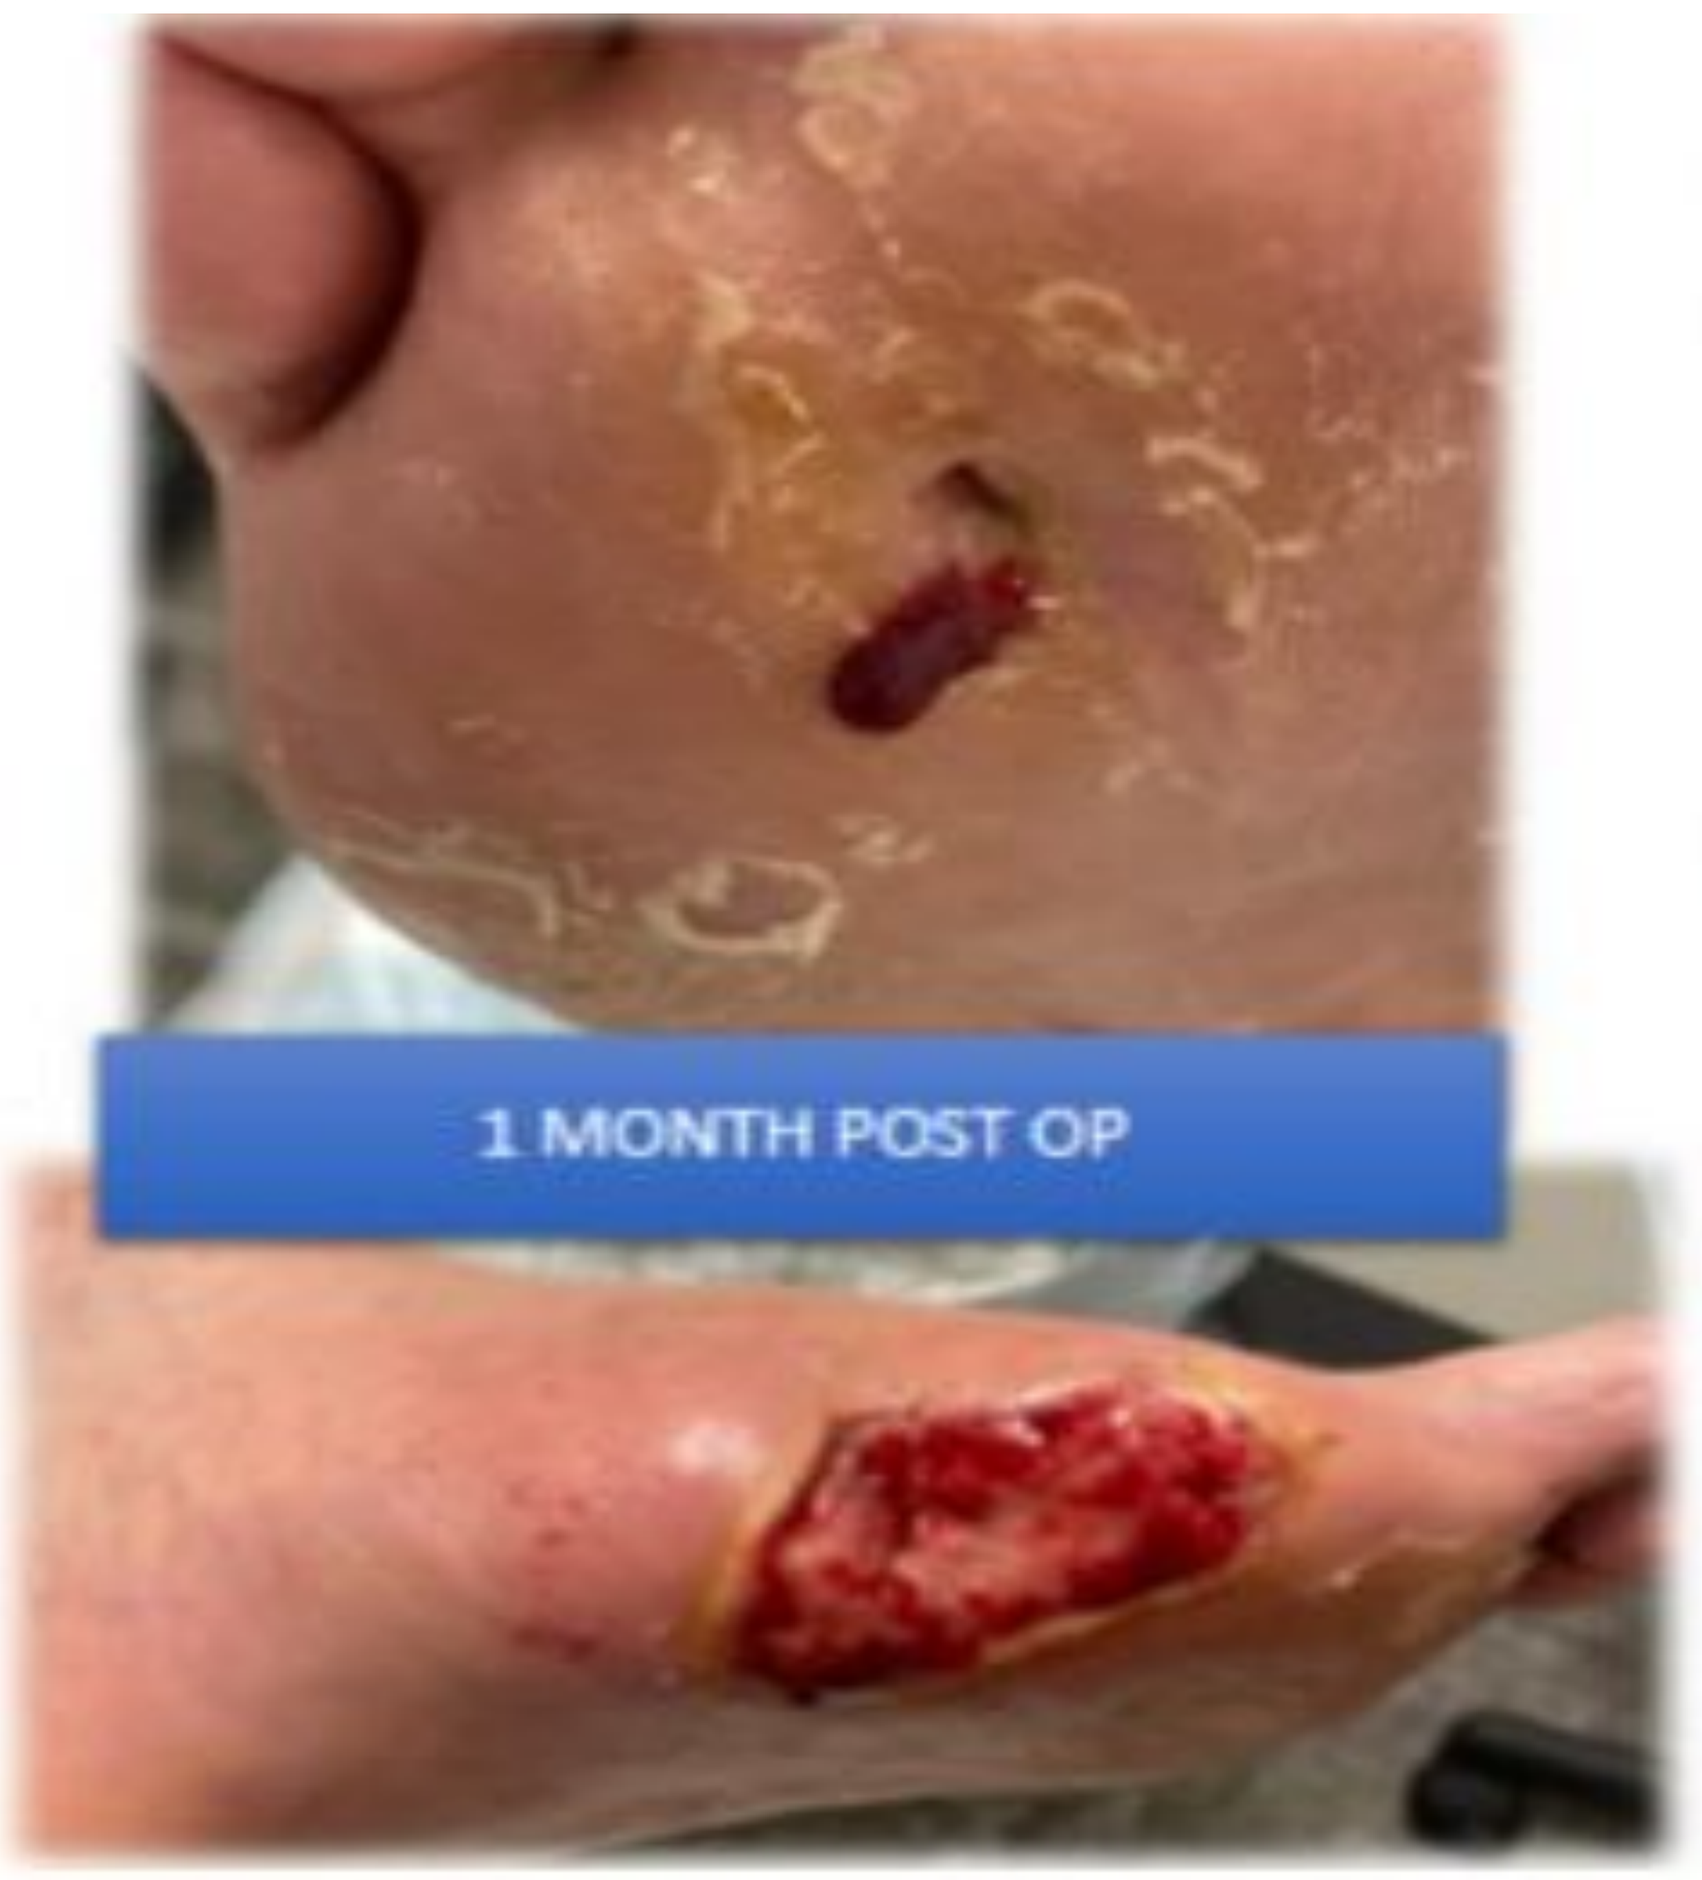
1 MONTH POST OP

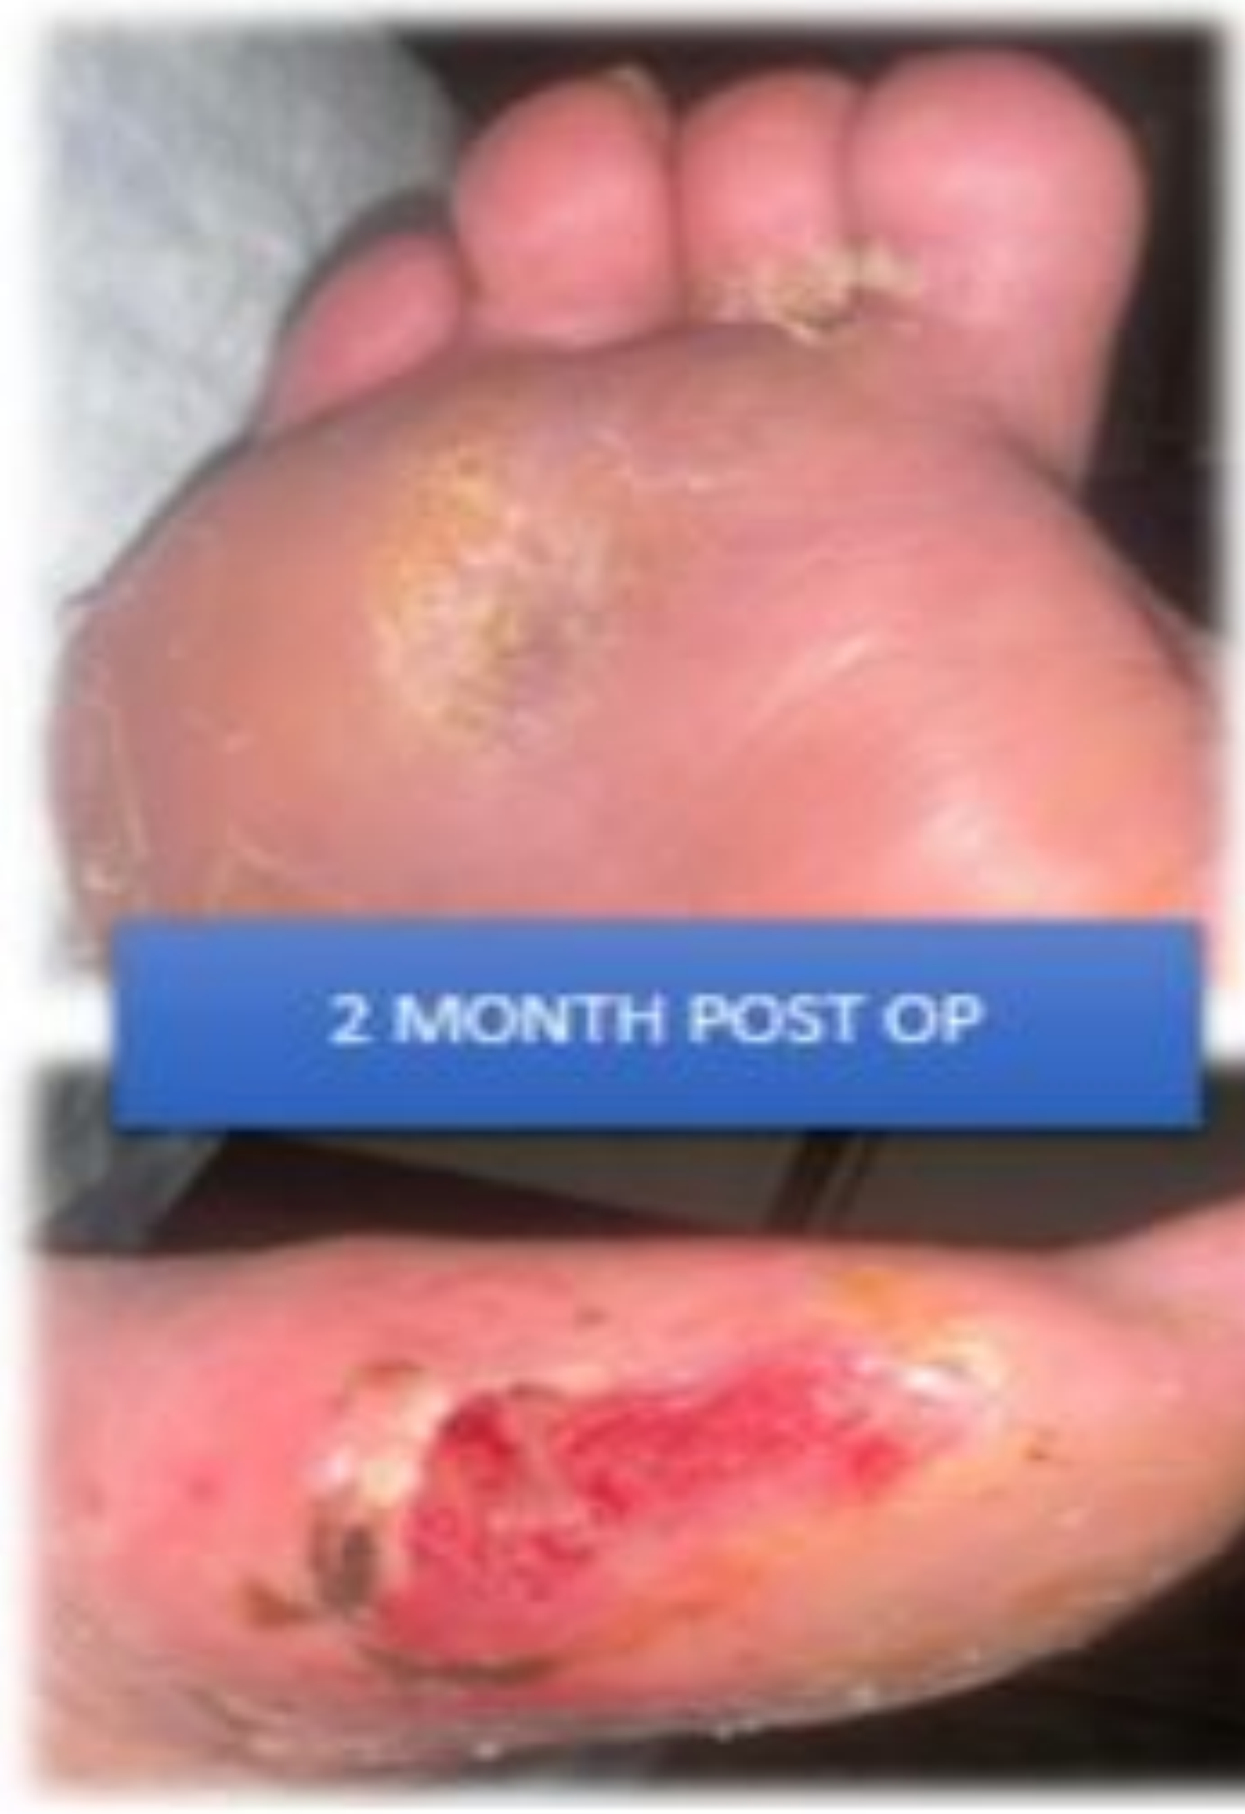
2 MONTH POST OP

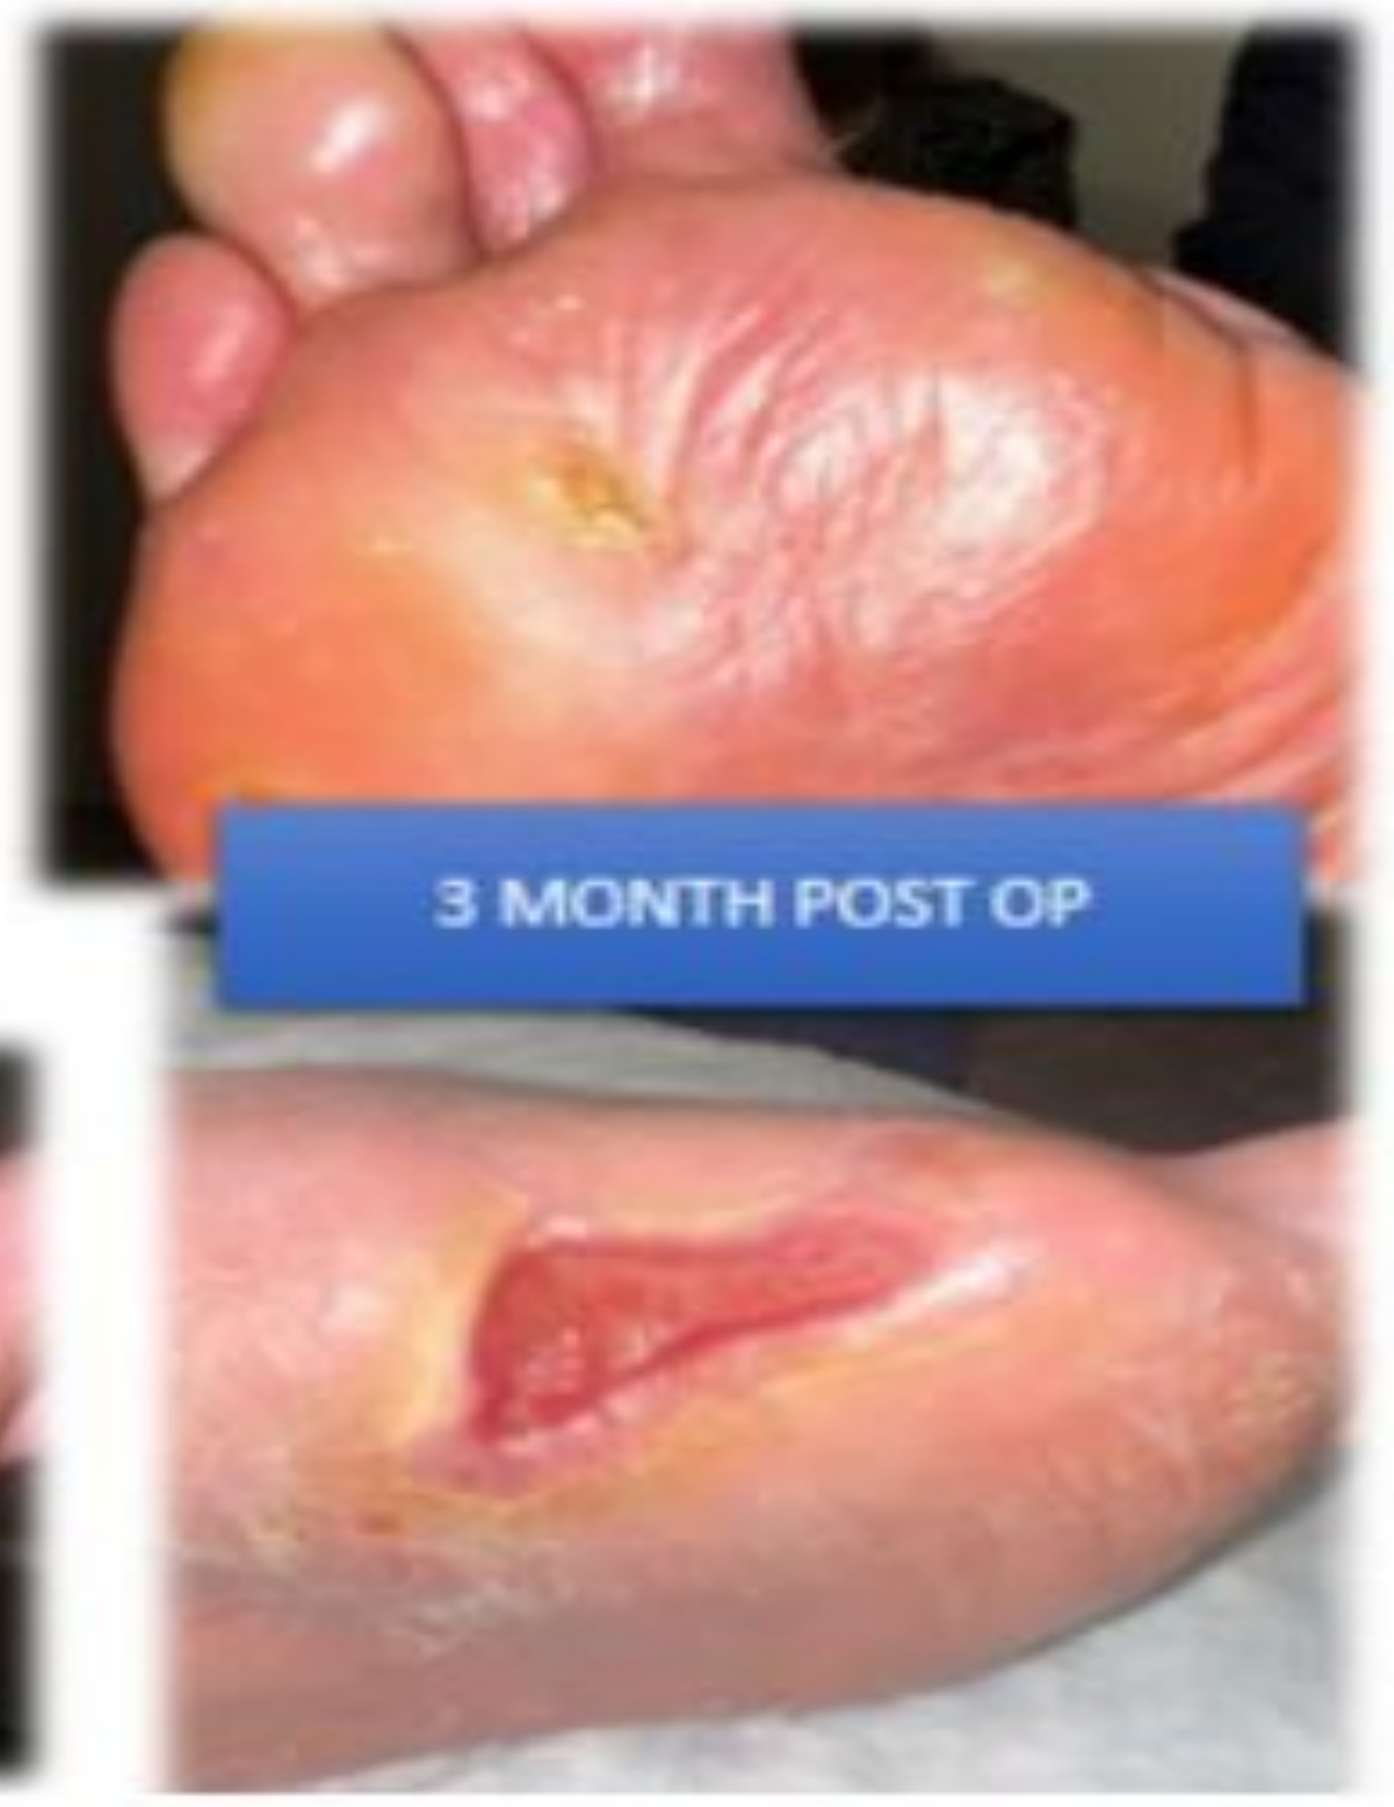
3 MONTH POST OP

Outcome

Stage 1: Left 1st and right third metatarsal resections

- Followed by thrombectomy and deployment of a percutaneous transvenous femoropopliteal bypass to the left leg.
- Wound vac therapy and surgical debridement were performed.

Stage 2: Two weeks later with TOT five times weekly for 1.5 hours/day.

- Complete healing of the right foot by month two
- Near-complete healing of the left foot by month four.

Stage 3: Right SFA stenting and angioplasty.

- TOT was completed,
- By six months, both ulcerations had fully resolved.
- The patient has remained under ongoing multidisciplinary follow-up, with no recurrence at one-year review.

Conclusion

This case illustrates the synergistic benefit of combining endovascular bypass with topical oxygen therapy in complex limb salvage. Through serial interventions and longitudinal management, the patient achieved complete bilateral wound healing within six months and sustained closure at one year, highlighting the potential of integrated perfusion and oxygen strategies in managing advanced vascular and diabetic foot ulcers

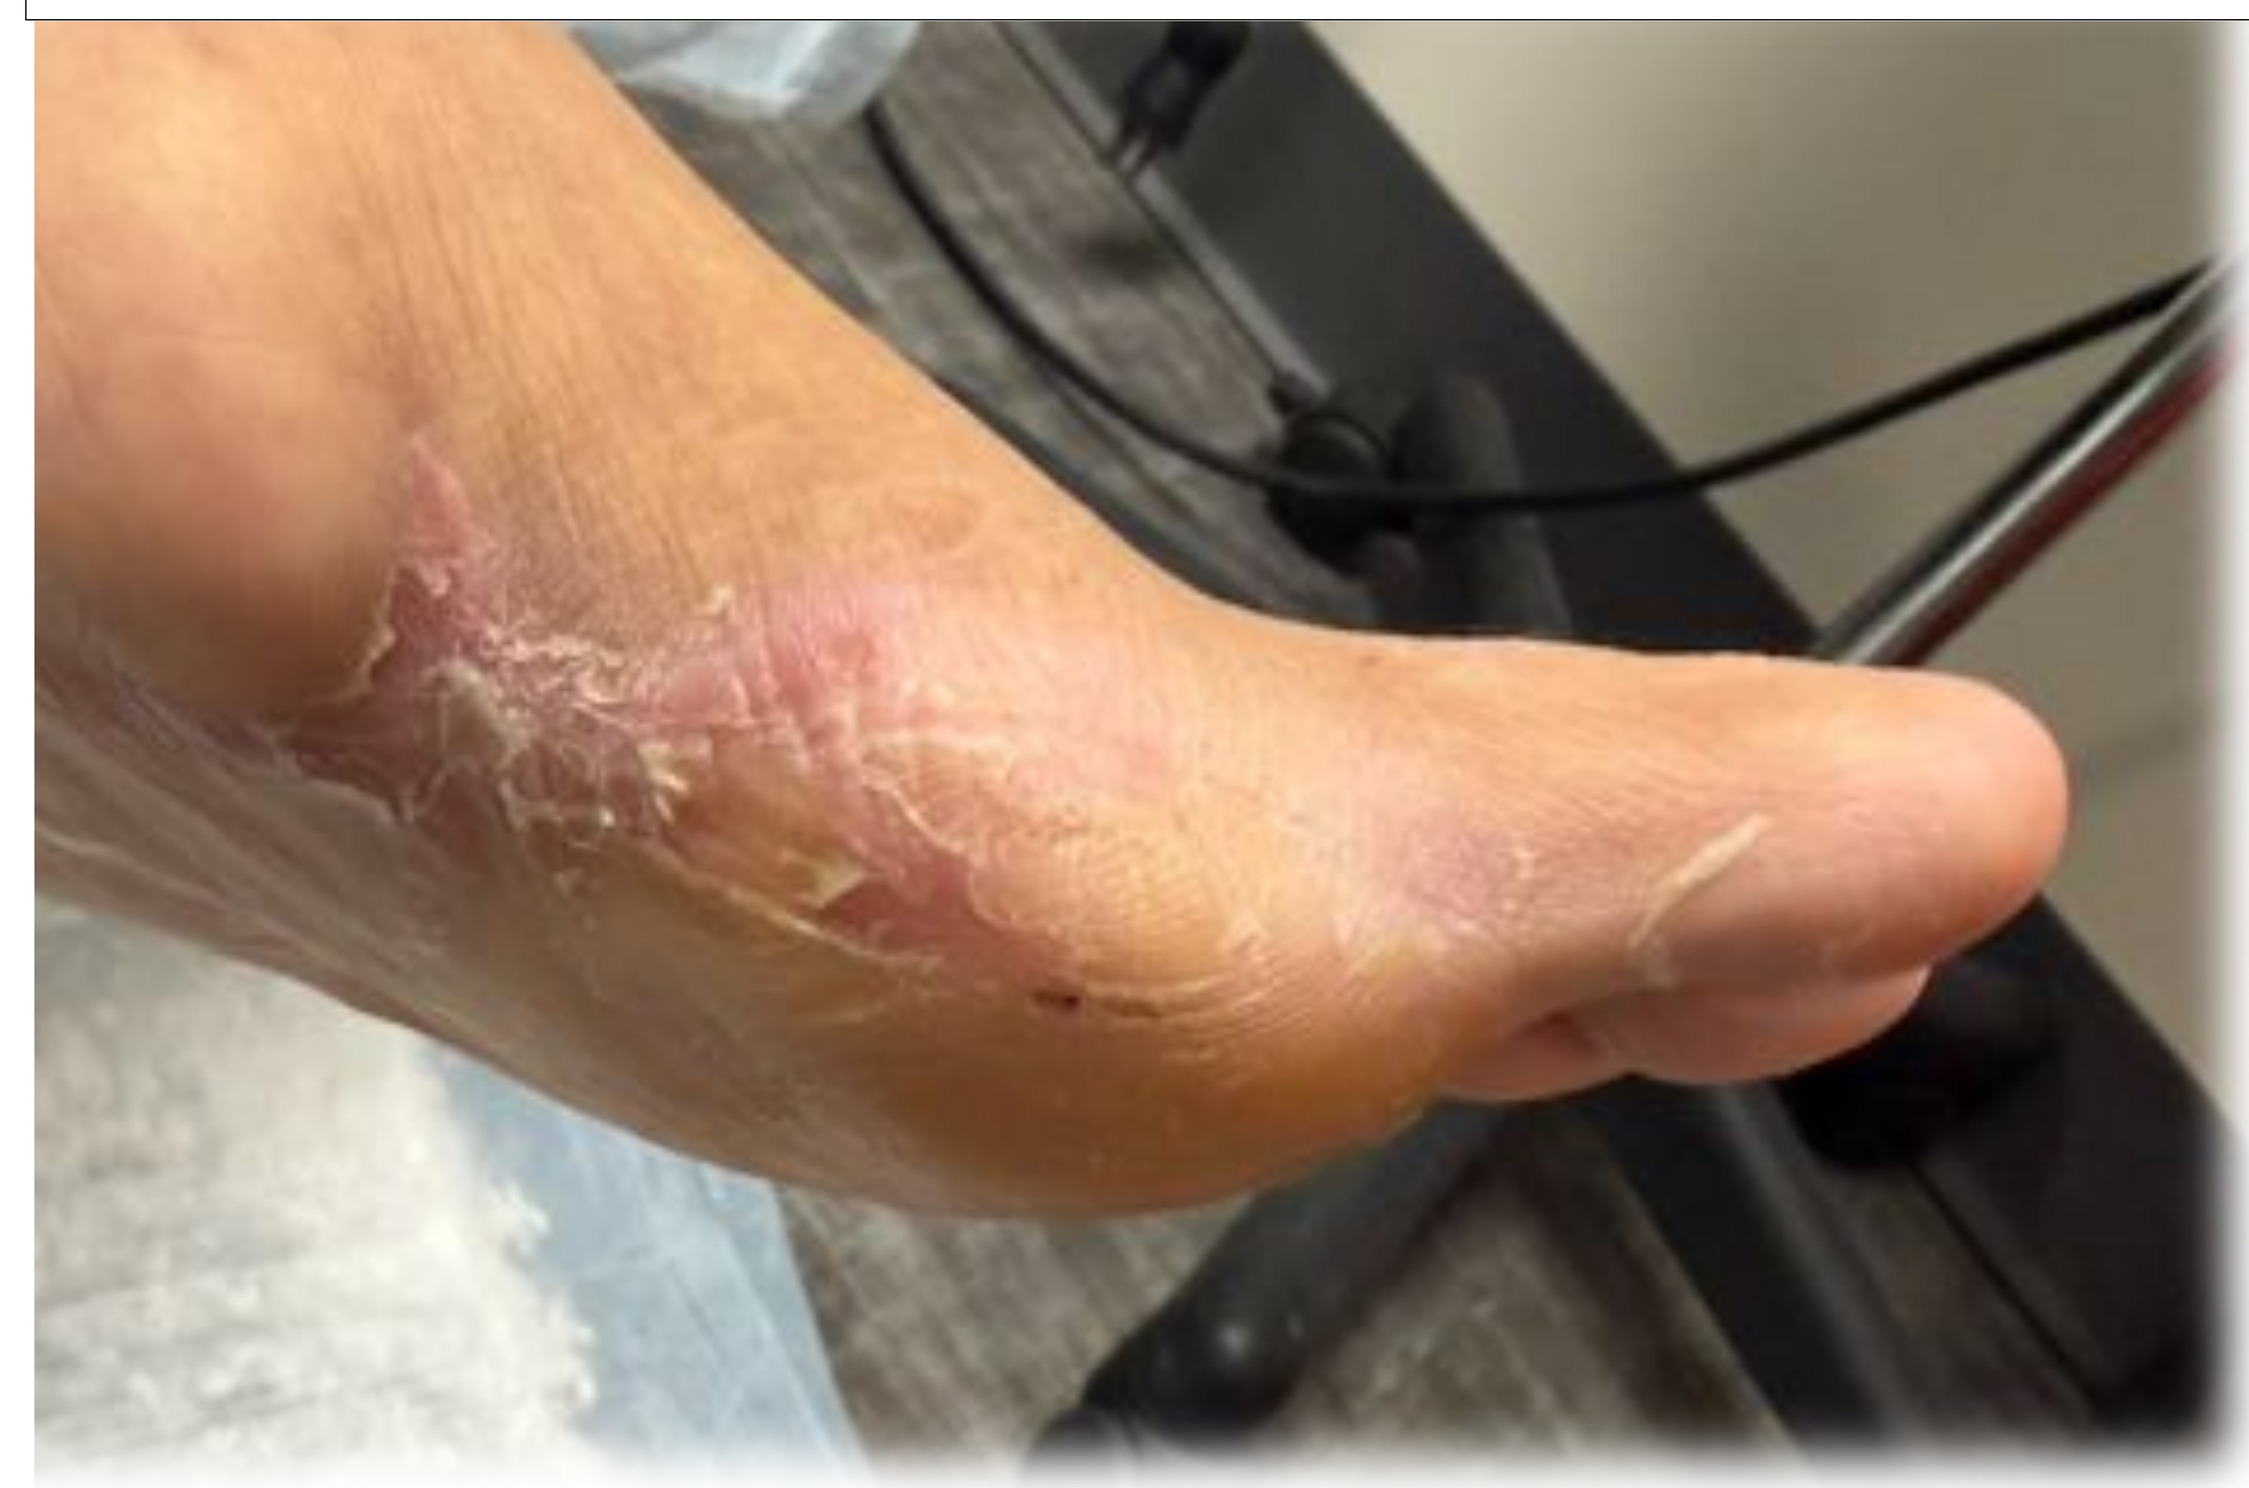

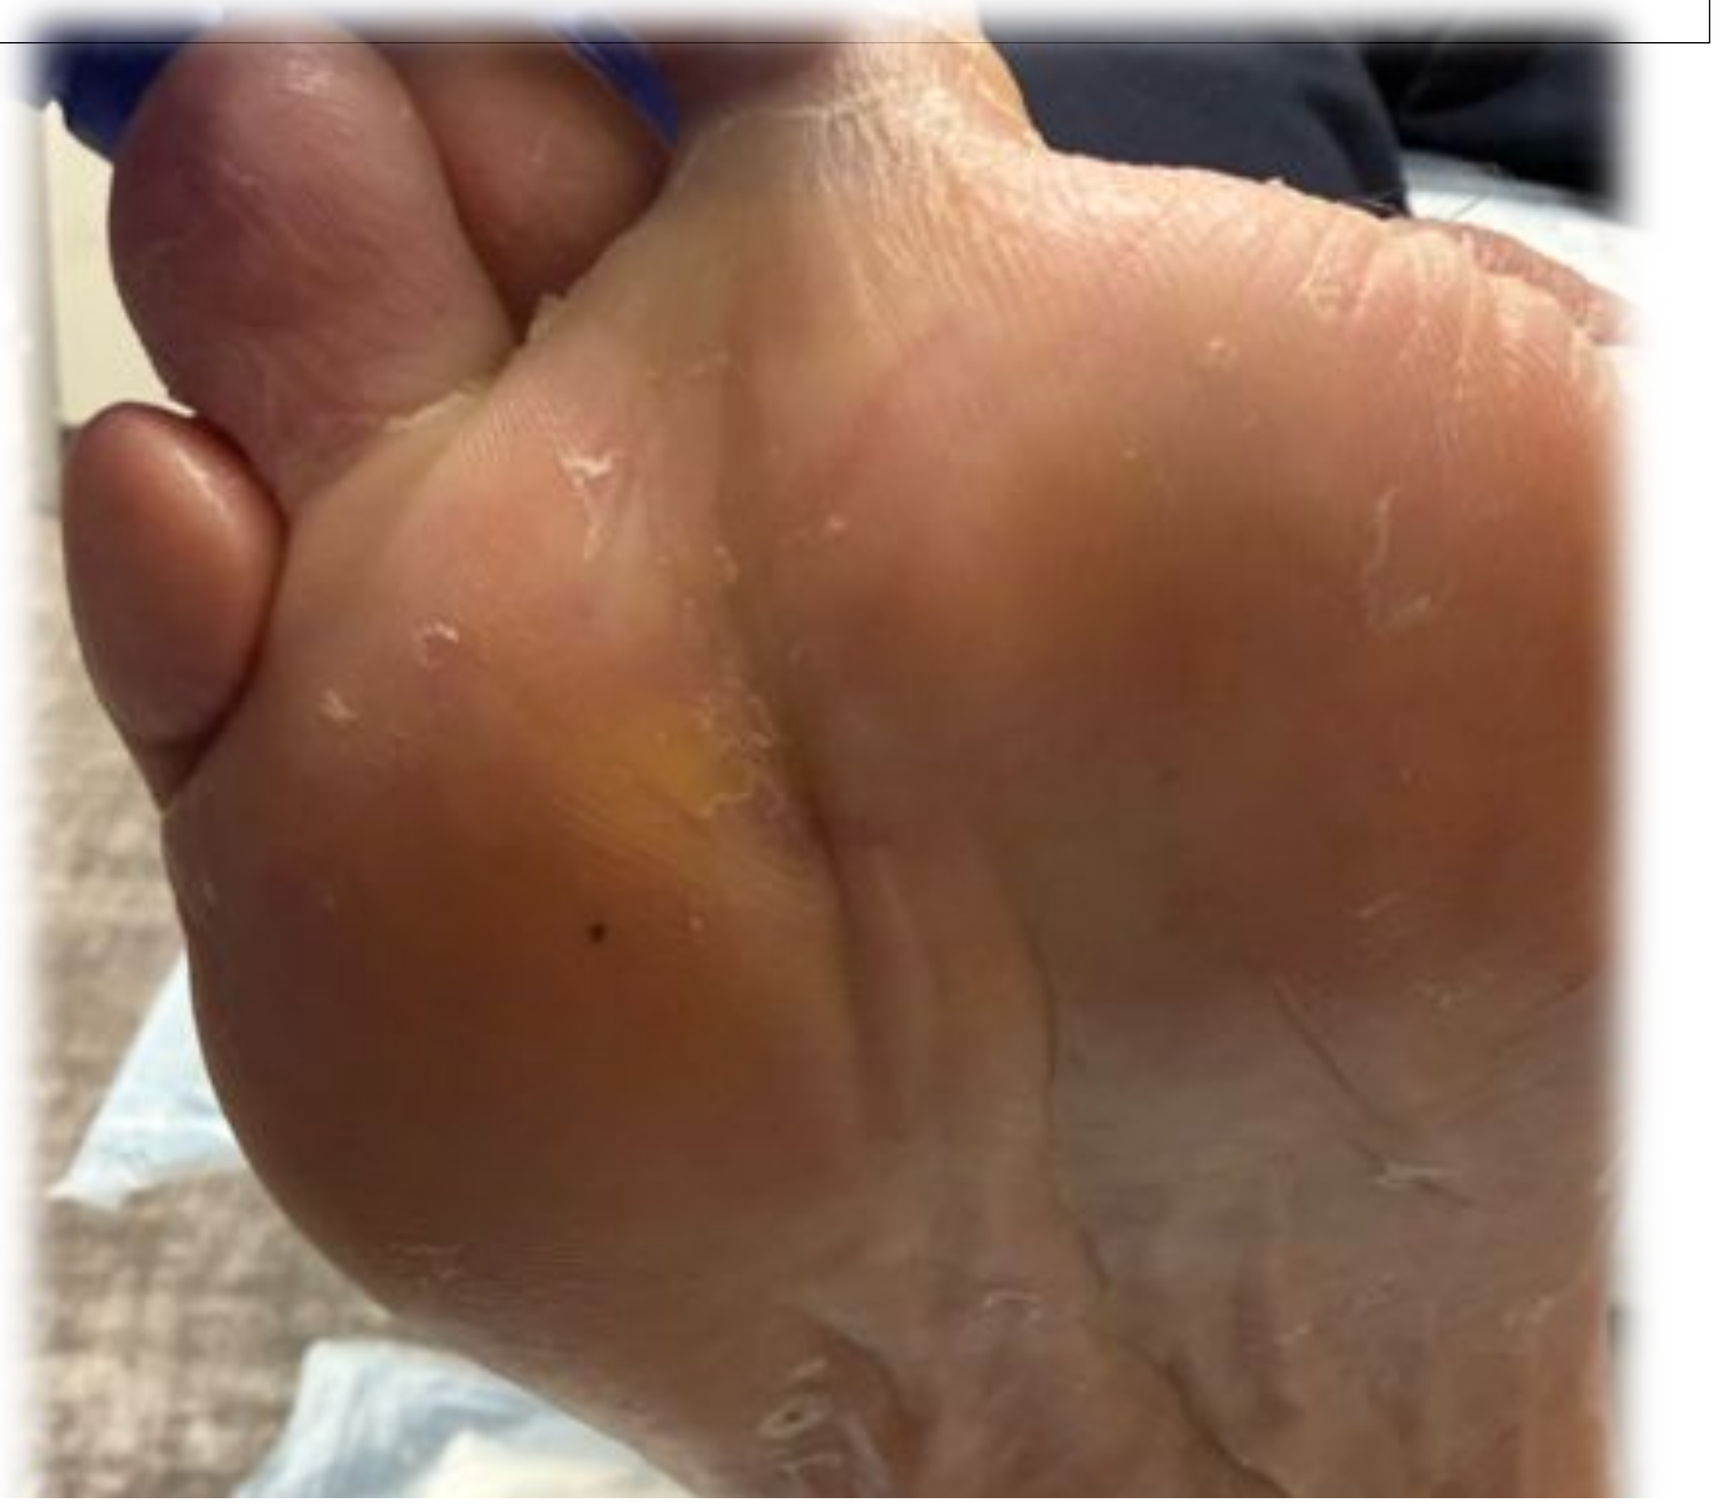

References

1. Dissemond J, Kröger K, Storck M, et al. Topical oxygen wound therapies: An international consensus. J Wound Care. 2023;32(Sup7):S1–S36. doi:10.12968/jowc.2023.32.Sup7.S1
2. Howard MA, Aslam R, Stojadinovic A, et al. Topical oxygen therapy in wound healing: A narrative review of mechanisms and modalities. Auctores Online. 2023. Accessed September 2025. https://auctoresonline.org/article/topical-oxygen-therapy-in-woundhealing-a-narrative-review-of-mechanisms-and-modalities
3. Hayes P, et al. Meta-analysis of topical oxygen therapy for chronic wounds. Int Wound J. 2024;21(3):456–468. doi:10.1111/iwj.14960
4. Cleveland Clinic. Pooled 2-Year Outcomes of DETOUR Trials Support Percutaneous Transfemoral Arterial Bypass. Consult QD. Published May 2024. Accessed September 2025. https://consultqd.clevelandclinic.org/pooled-2-year-outcomes-of-detour-trialssupport-percutaneous-transfemoral-arterial-bypass
5. Endologix Inc. DETOUR2 Trial Final Results Evaluate Endologix DETOUR for PTAB to Treat Complex Femoropopliteal Disease. Endovascular Today. Published March 2025. Accessed September 2025. https://evtoday.com/news/detour2-final-results-evaluateendologix-detour-for-ptab-to-treat-complex-femoropopliteal-diseas