Ulcer Unplugged: How Detour Bypass and Oxygen Therapy Accelerate Repair

Hayley Collins DPM, William Lopez DPM, Mitchell Weinberg MD
Staten Island University Hospital; Staten Island NY

Purpose

To demonstrate the synergistic impact of combining topical oxygen therapy (TOT) with percutaneous transvenous femoropopliteal bypass in the longitudinal management of complex diabetic foot ulcers and advanced peripheral arterial disease.

Background

Topical oxygen therapy (TOT) is an emerging adjunct in the treatment of chronic wounds, particularly those complicated by vascular insufficiency. Oxygen supports key phases of healing—angiogenesis, collagen synthesis, epithelialization, and immune defense. A 2024 meta-analysis demonstrated that TOT significantly improves healing rates compared to standard care, with relative risks of 1.77 in randomized trials and 2.15 in observational studies. Advances in delivery systems now allow for continuous or cyclical oxygen application, enhancing outcomes in hypoxic wound beds. International consensus guidelines endorse TOT as part of multidisciplinary wound care, emphasizing appropriate patient selection.

Percutaneous transvenous femoropopliteal bypass offers a novel endovascular solution for long-segment superficial femoral artery (SFA) disease, particularly in cases of chronic total occlusion or in-stent restenosis. Unlike traditional open bypass, this technique uses a minimally invasive route to achieve durable revascularization. Pooled data from multicenter trials (n=273) demonstrated high technical success, low adverse event rates, and sustained patency. Final 3 year results reported primary patency of 58.2%, freedom from target lesion revascularization at 66.8%, and a major amputation rate of just 1.5%.

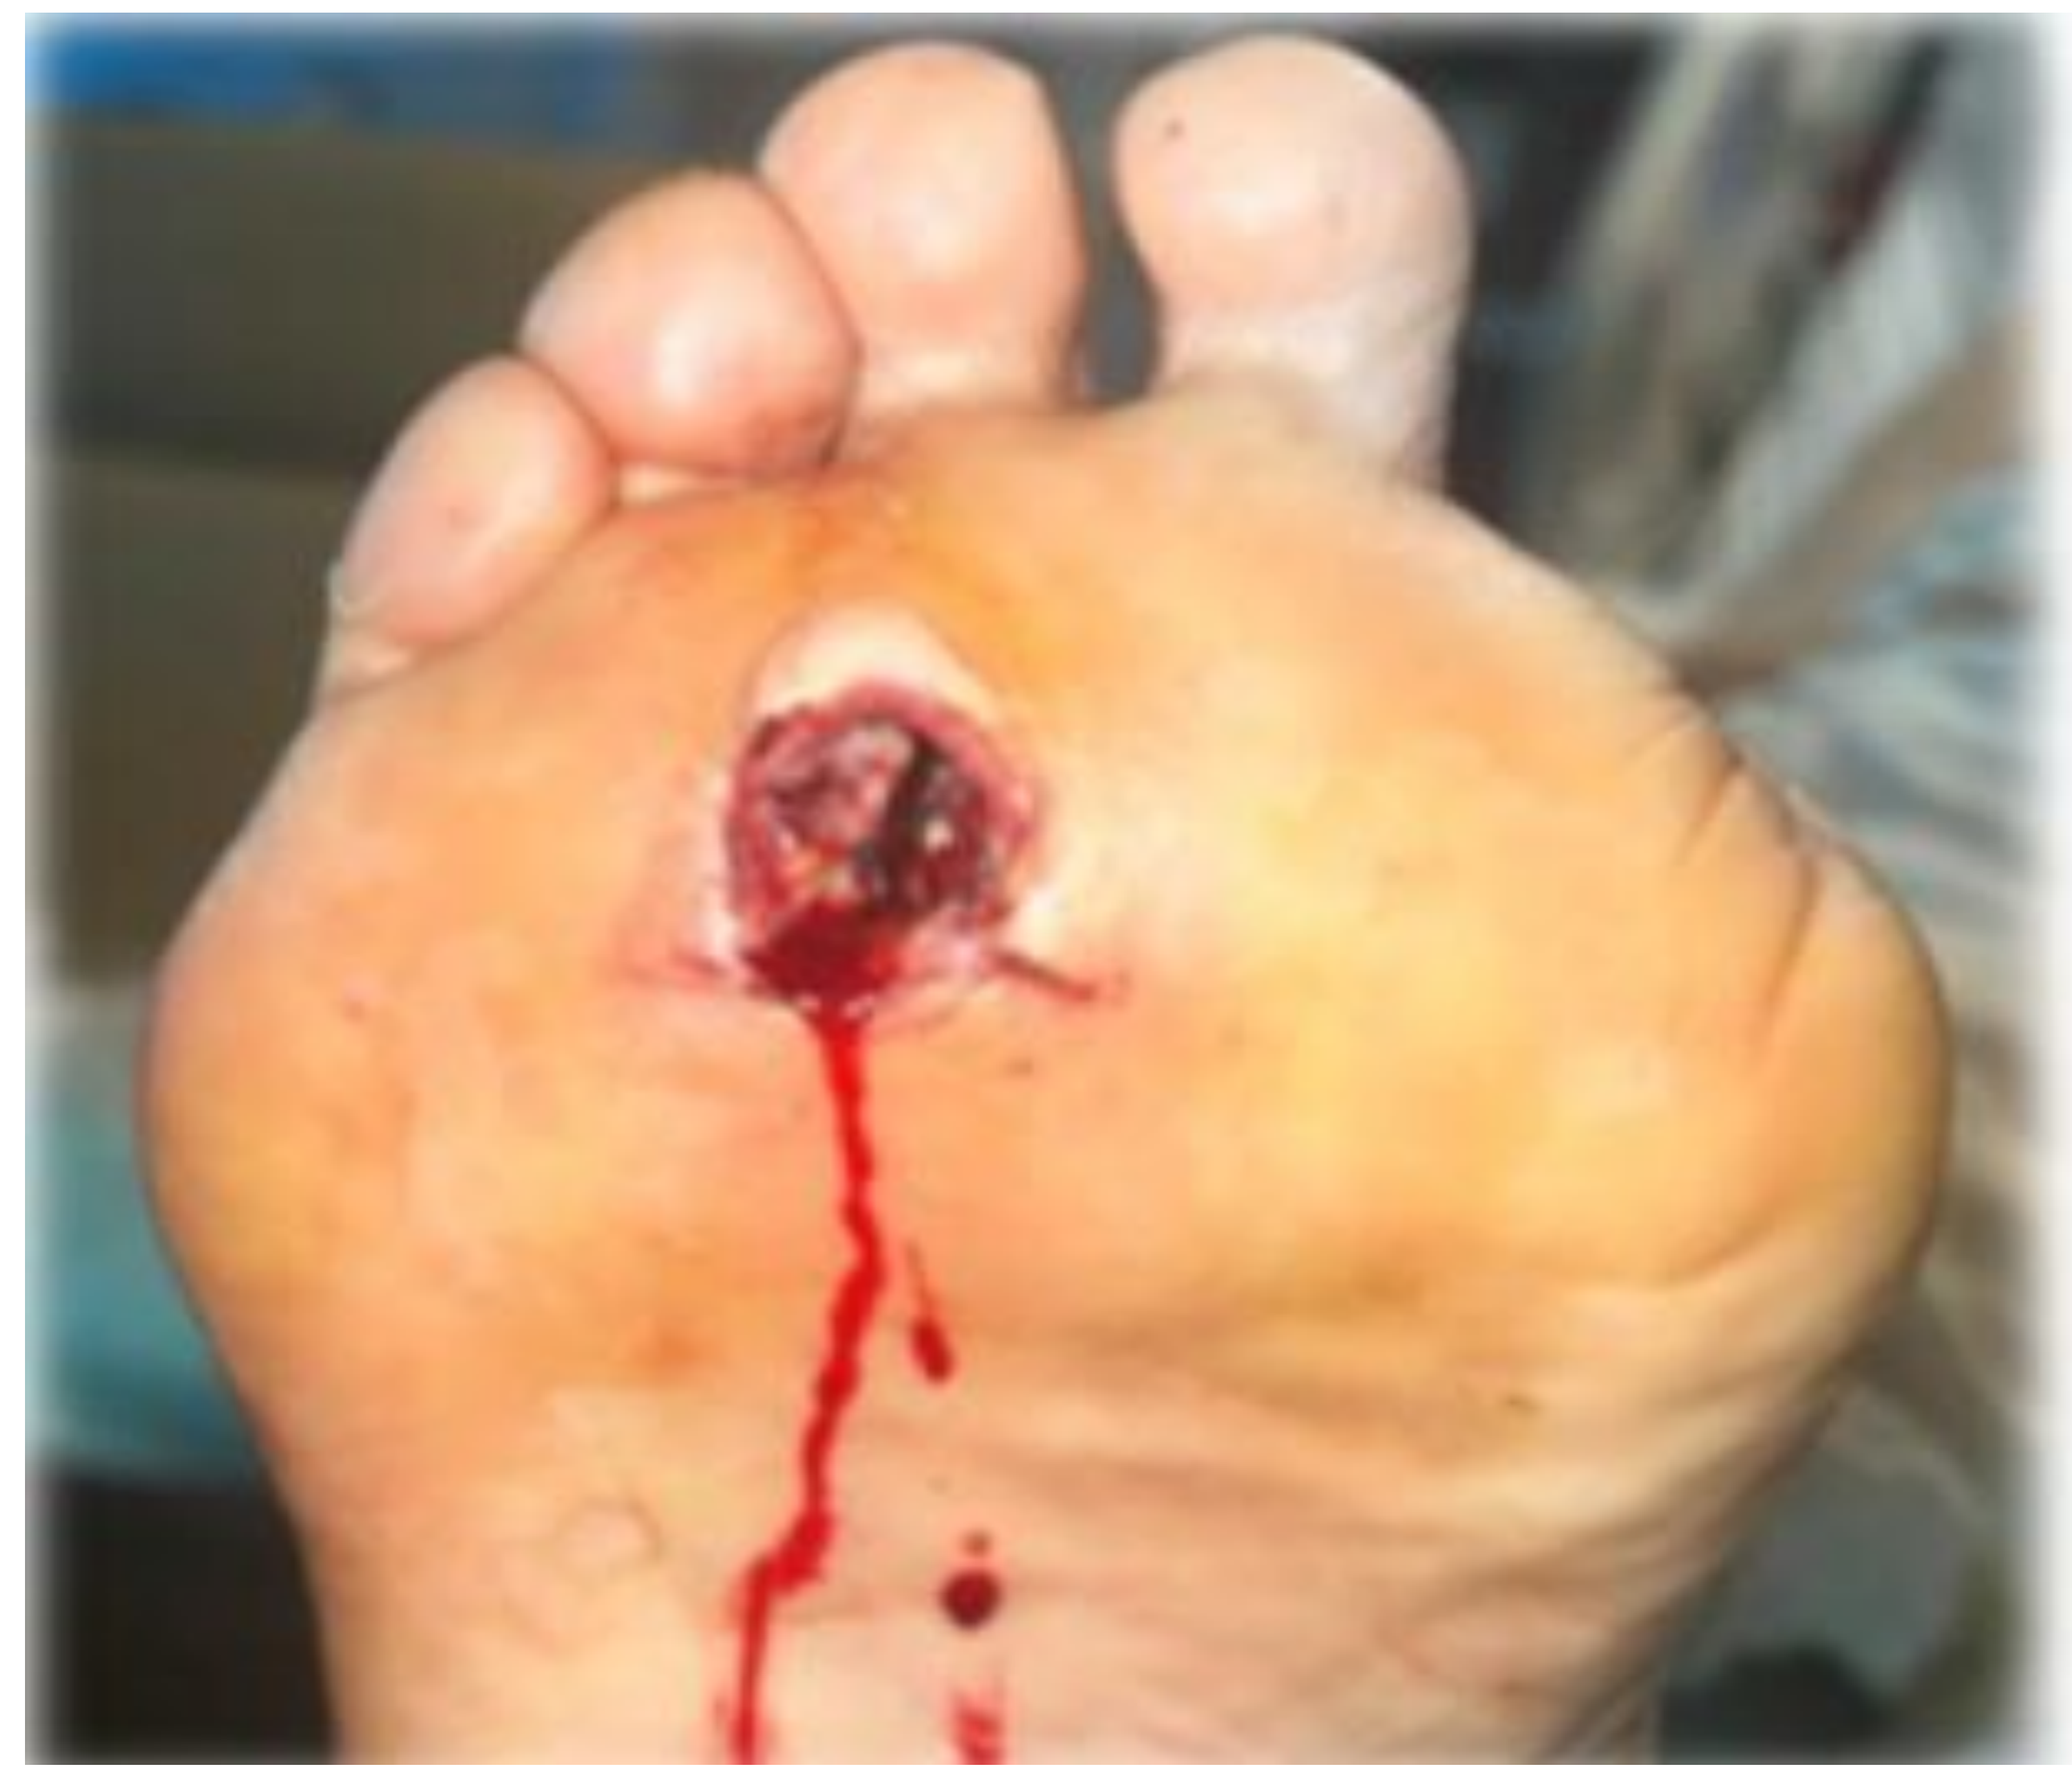

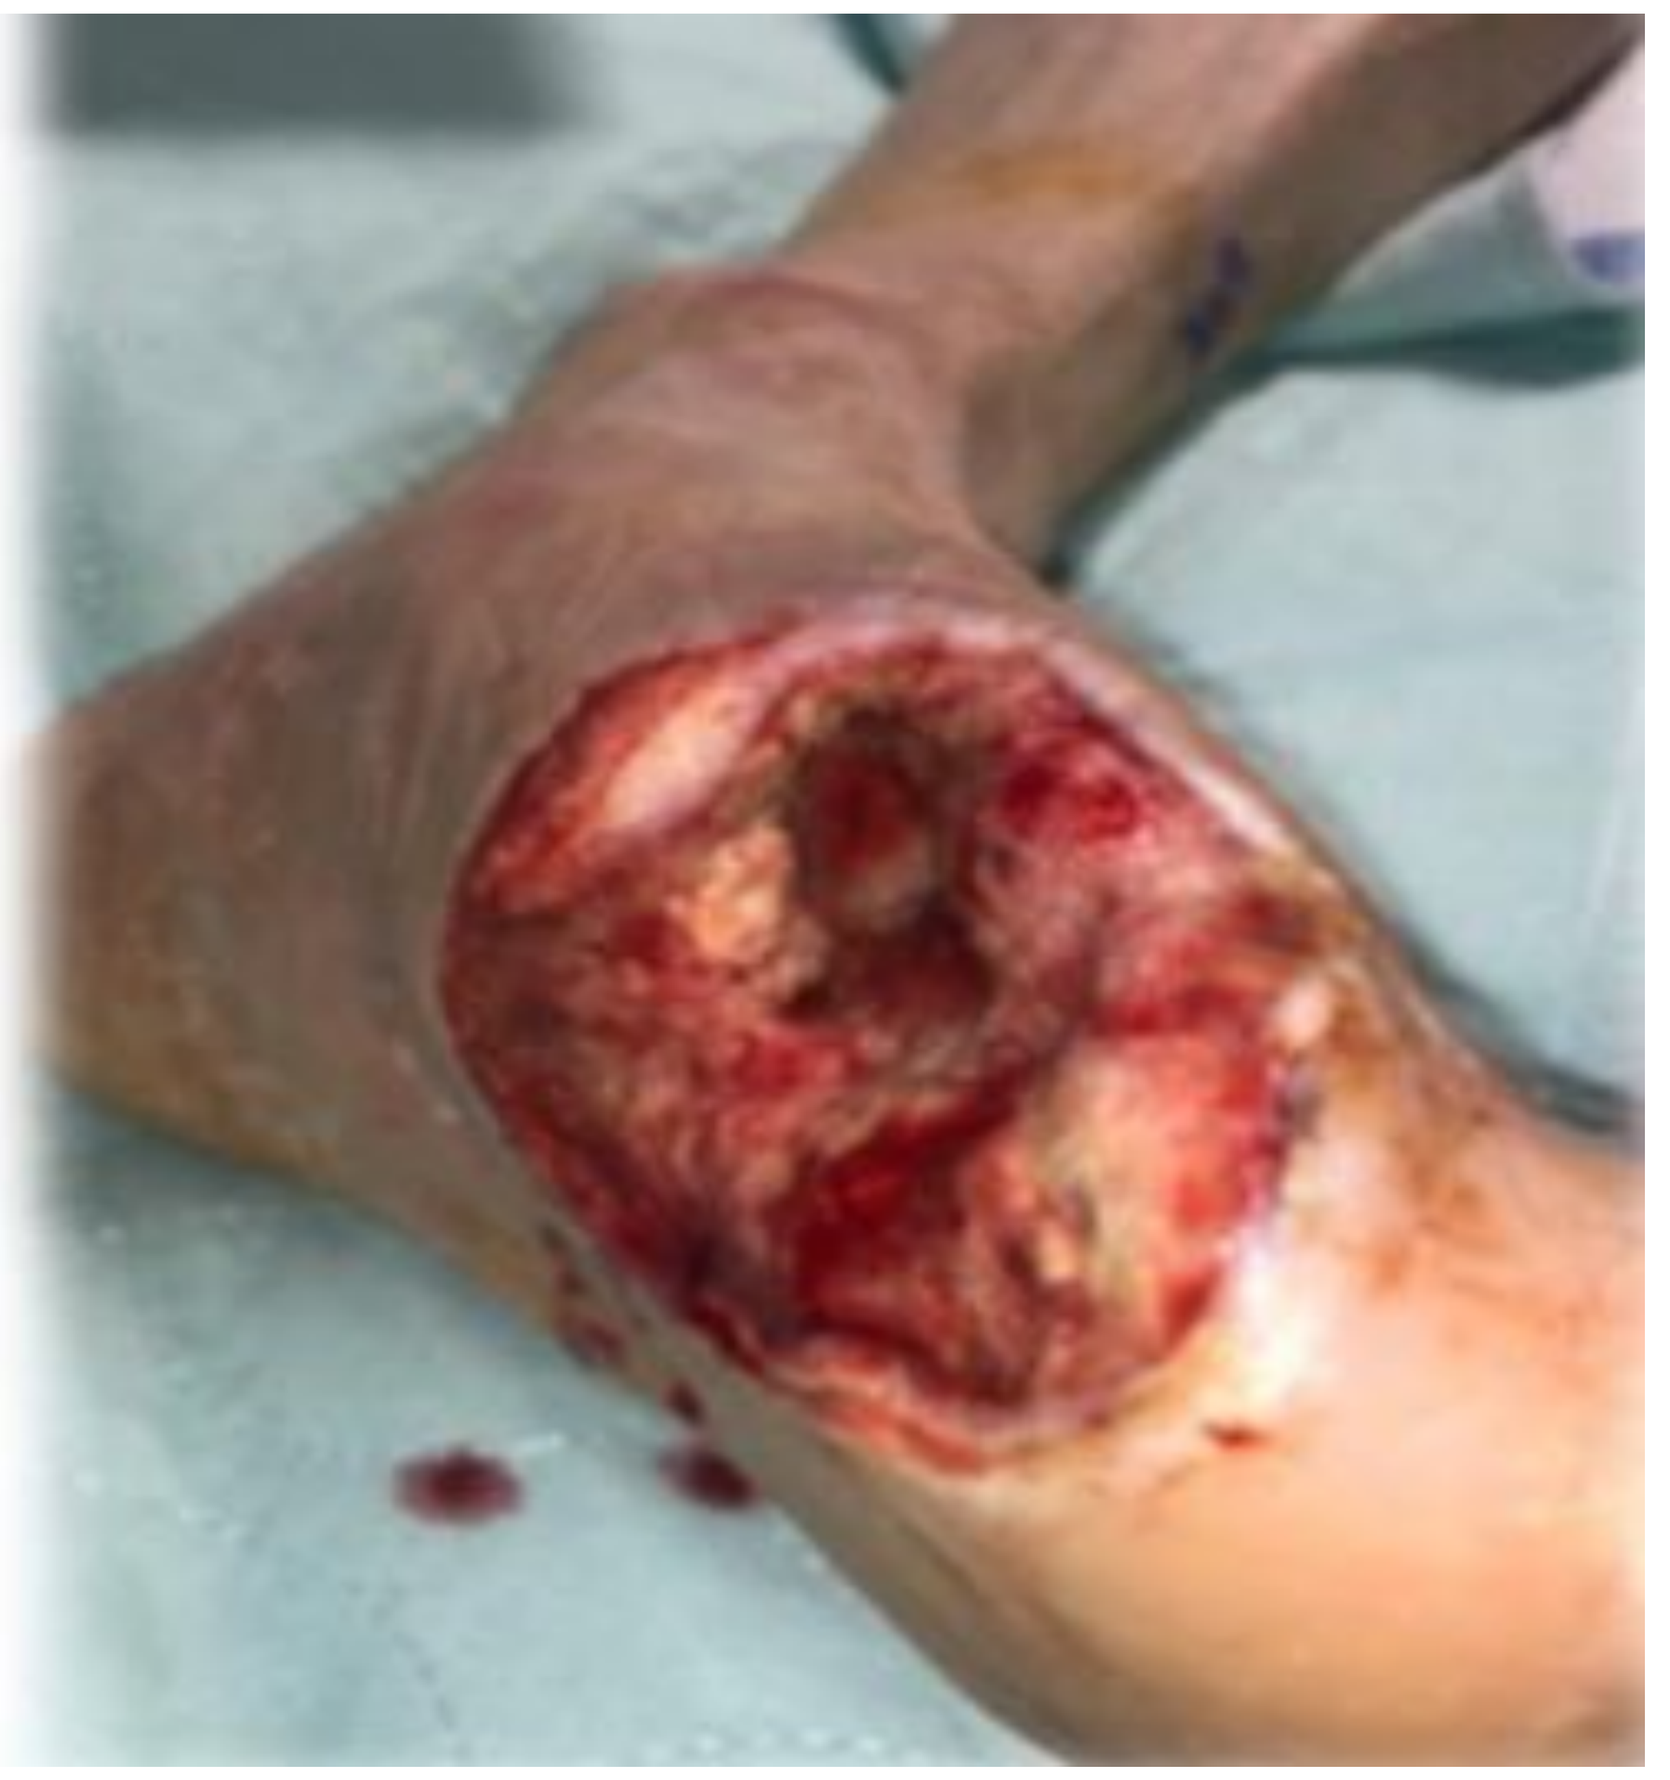

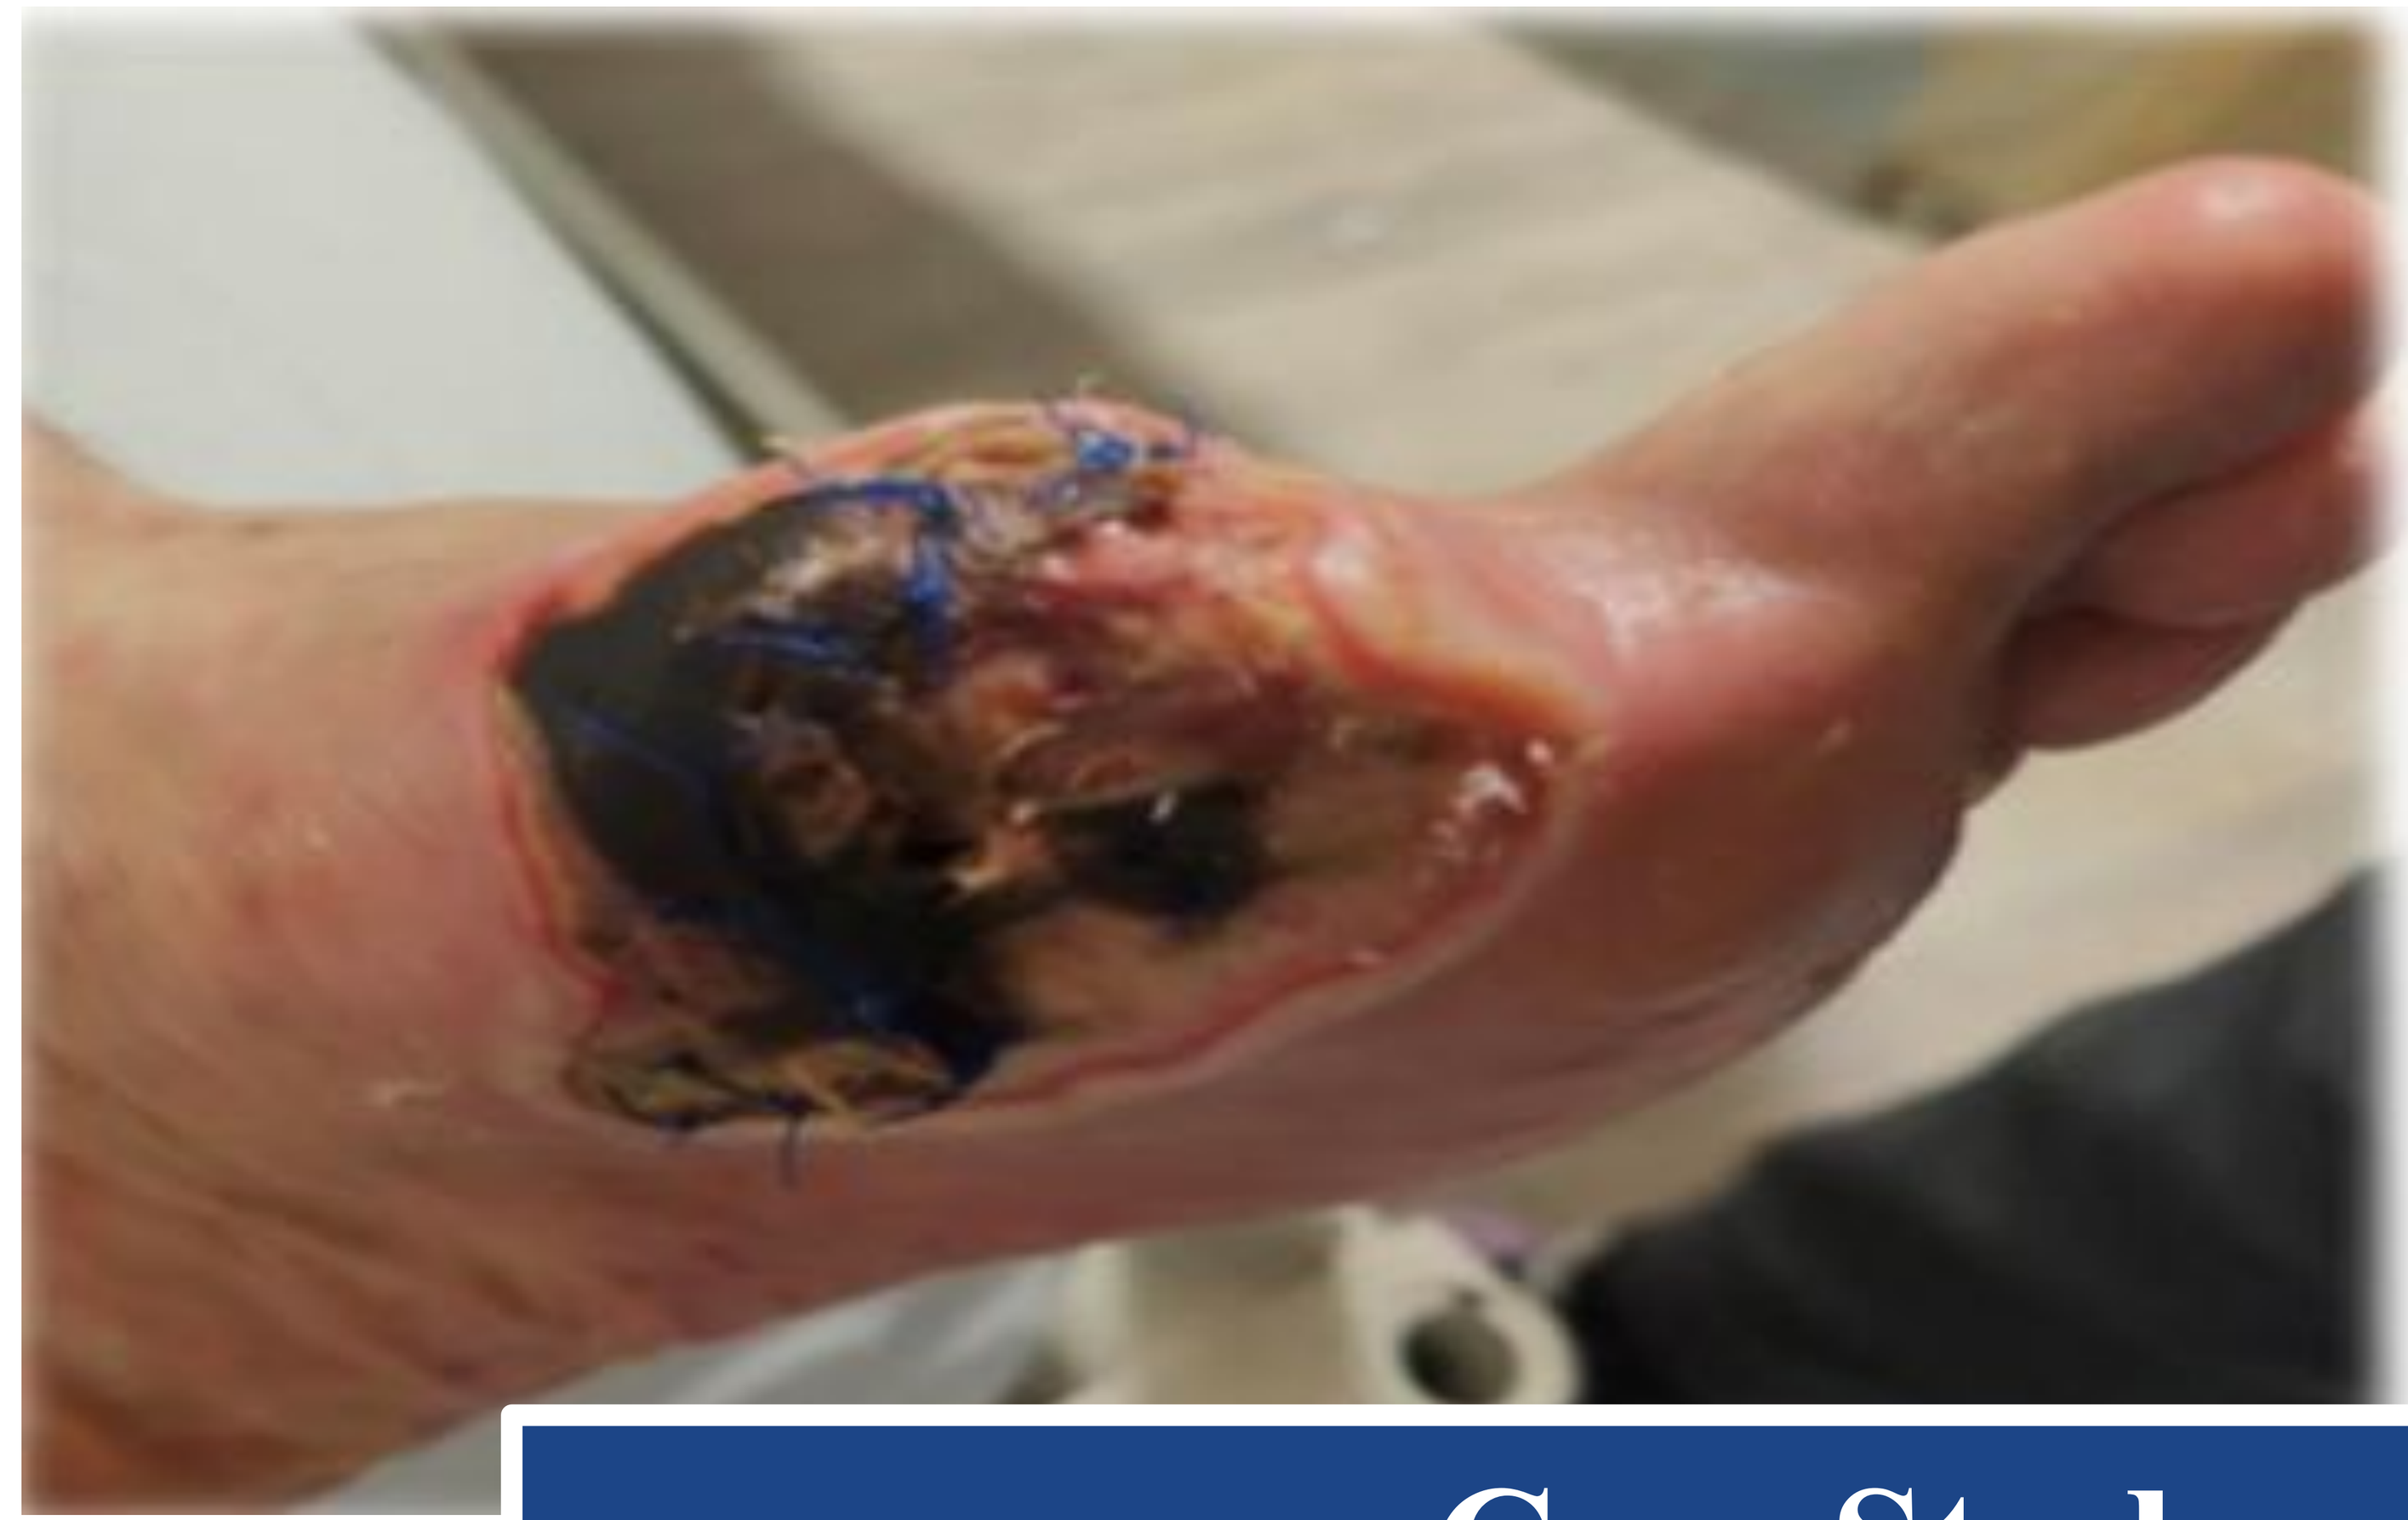

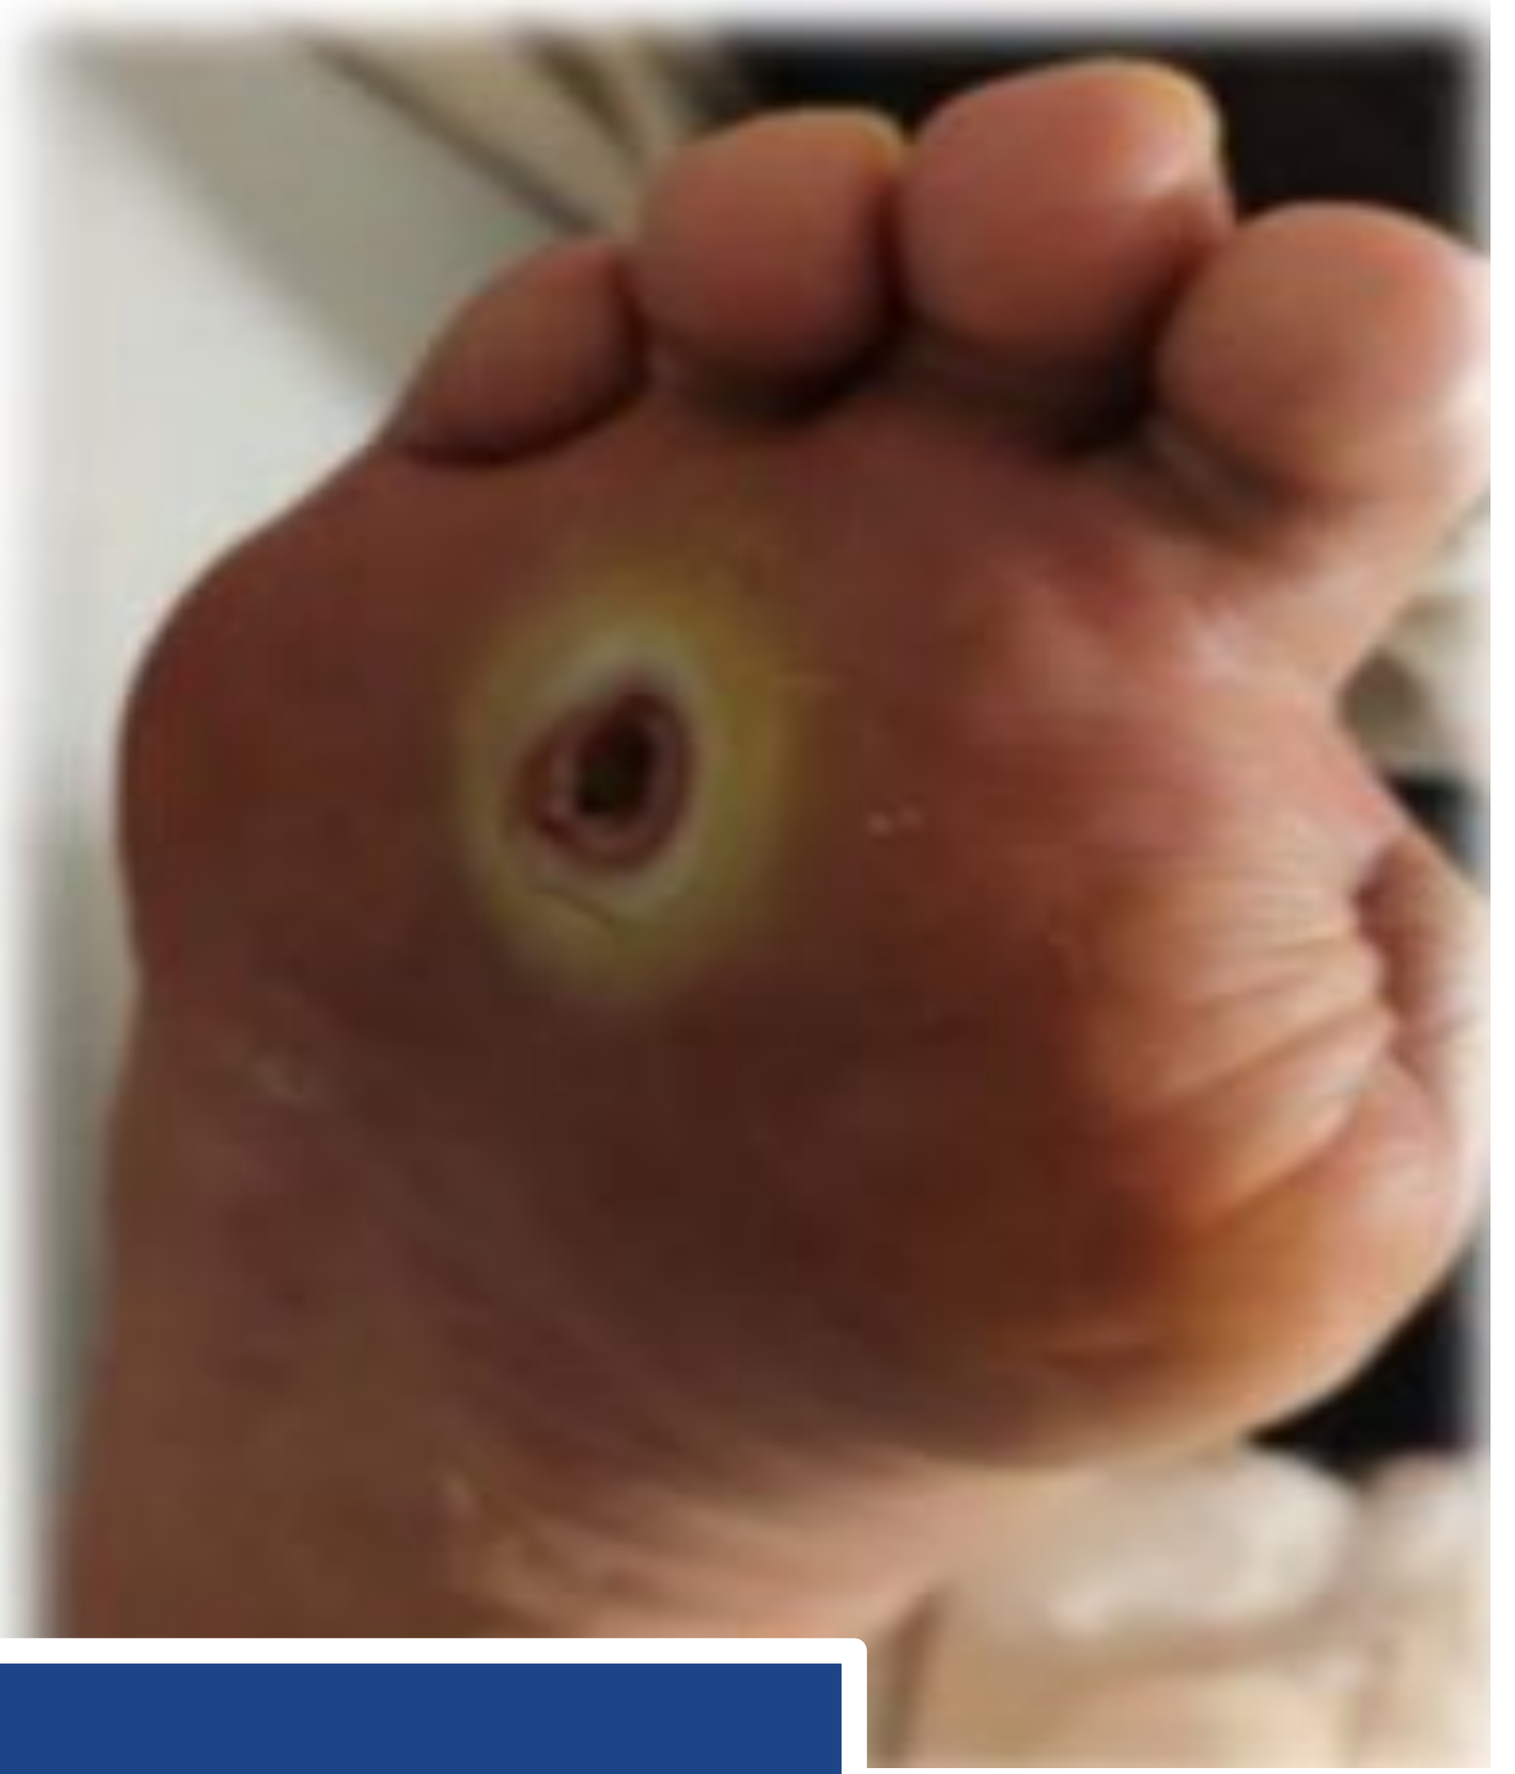

Case Study

A 68-year-old male with HTN, DM, HLD, and CAD has been followed for progressive PAD and recurrent ulcerations. Over the past year, he developed non-healing wounds and worsening claudication. Wound dehiscence was noted at the left hallux amputation site, with avascular necrosis and fibrotic base. A second ulcer under the right third metatarsal head showed purulent drainage and a sinus tract with probe-to-bone.

His vascular history included prior left-sided revascularization of the SFA, popliteal, and anterior tibial arteries, and PTA with three coronary DES to the right anterior tibial artery. Additional interventions included laser atherectomy and DCB to the left SFA and profunda. In 2024, he developed 90% stenosis of the right SFA and ISR of the left SFA/POP segment.

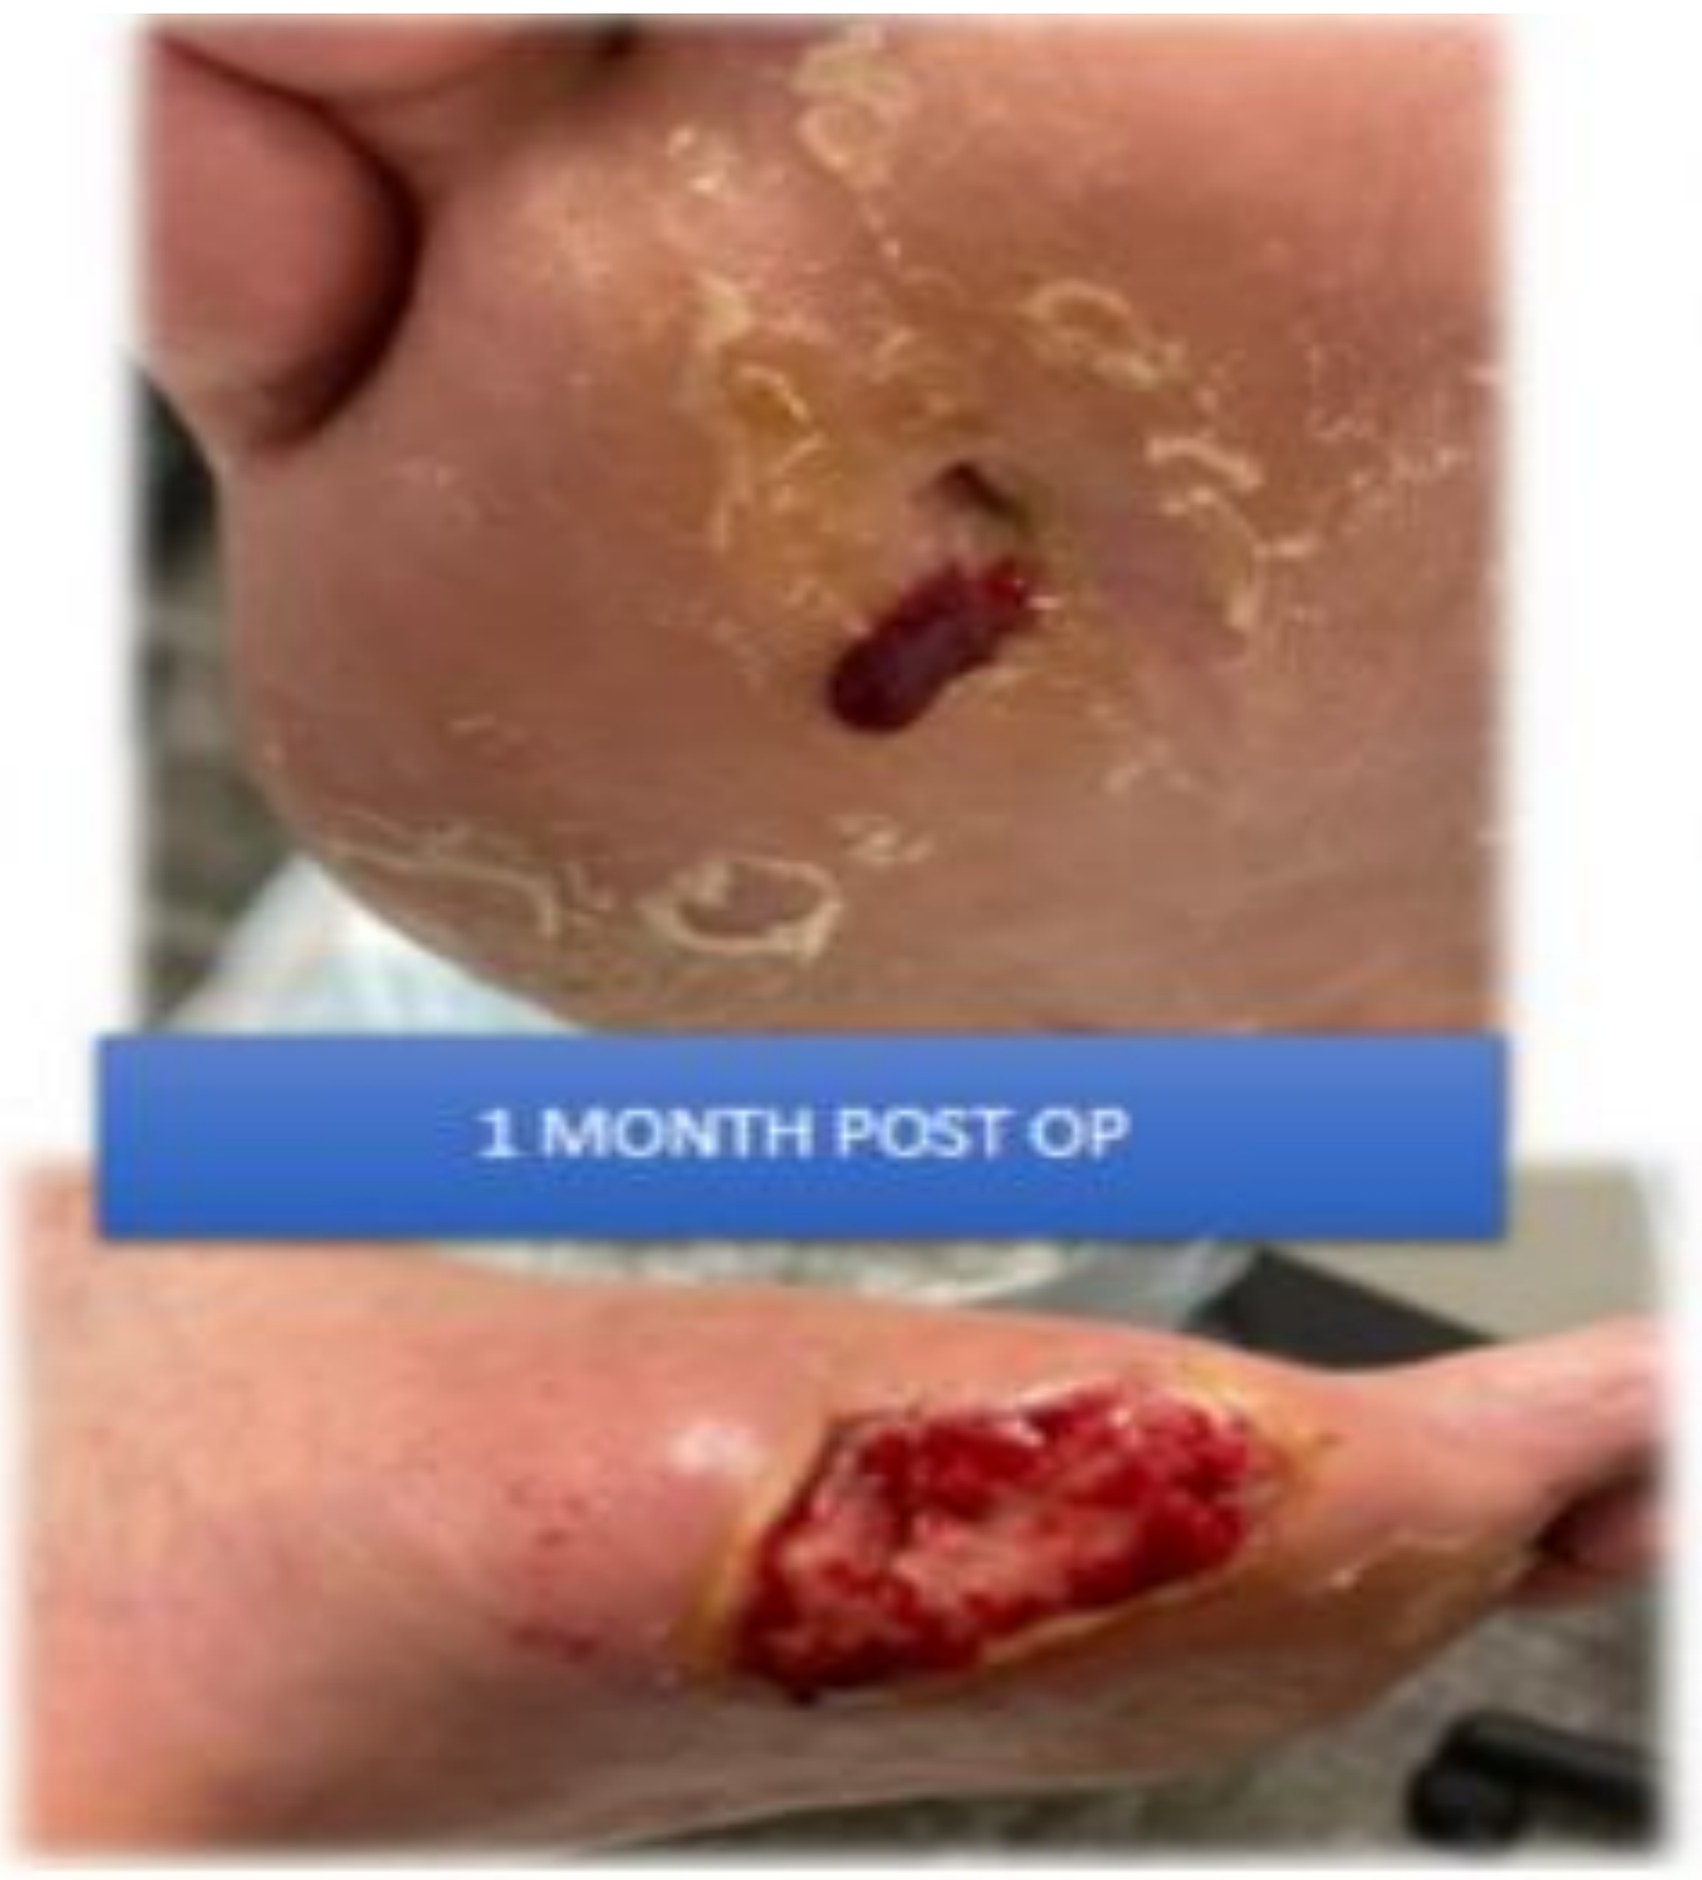
1 MONTH POST OP

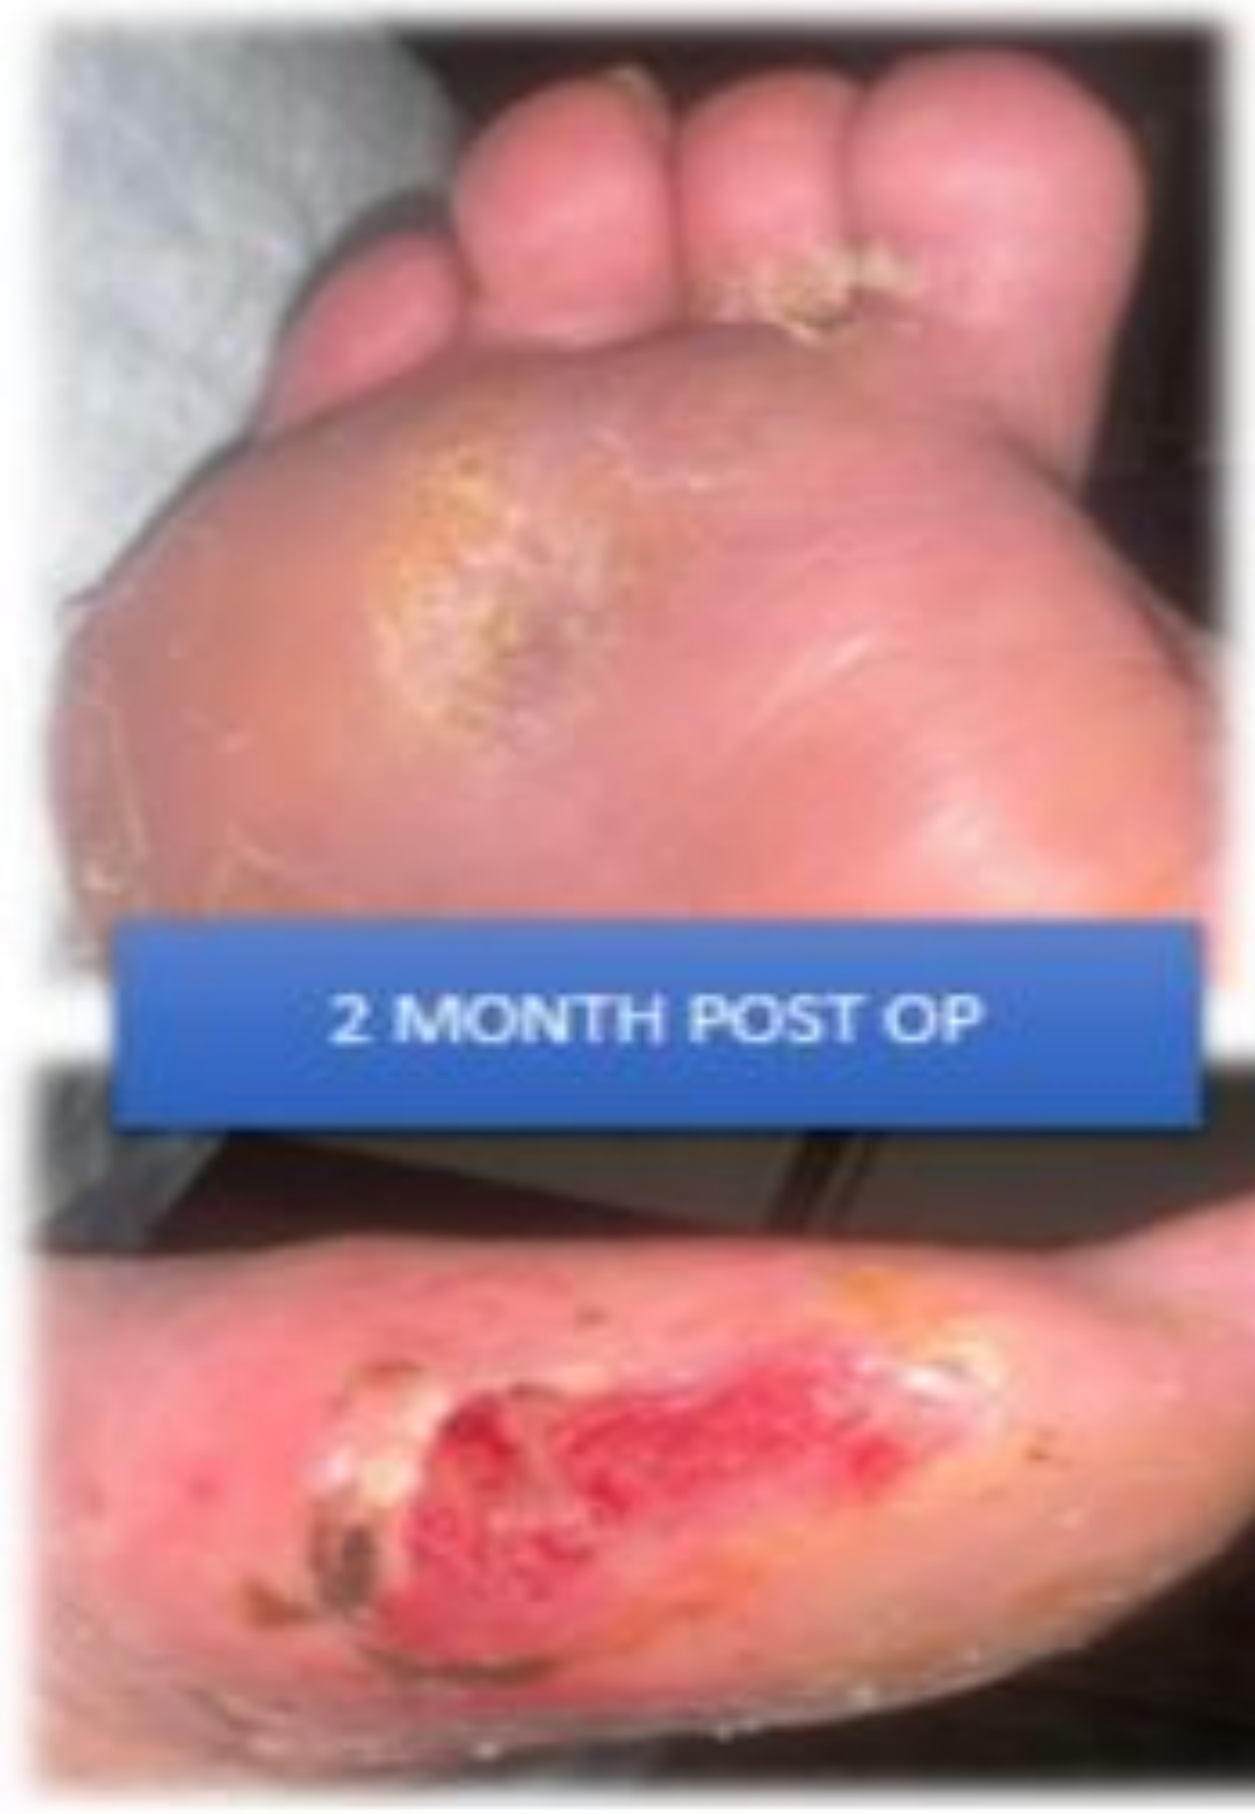
2 MONTH POST OP

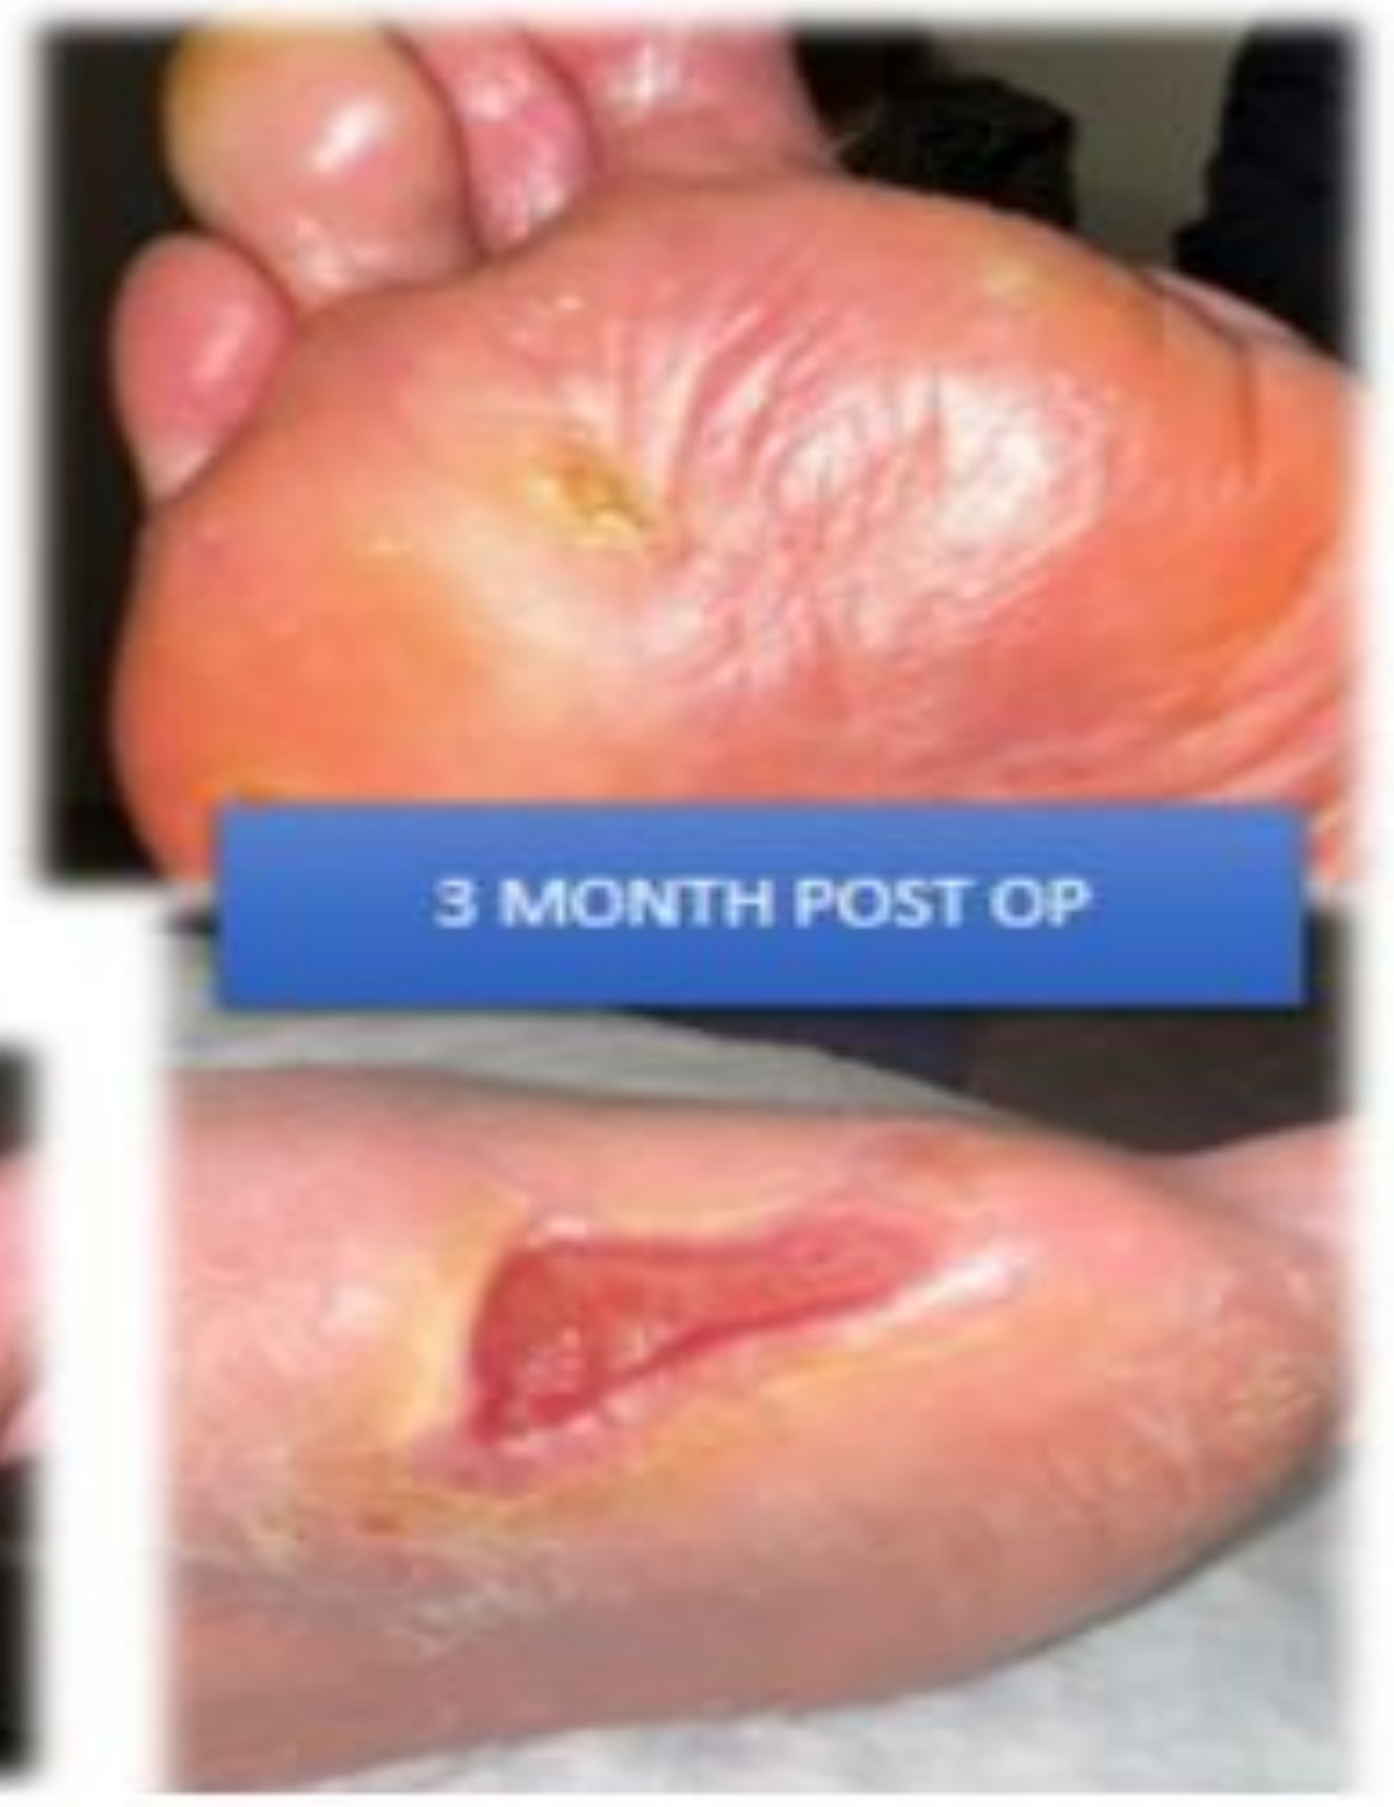
3 MONTH POST OP

Outcome

Stage 1: Left 1st and right third metatarsal resections
- Followed by thrombectomy and deployment of a percutaneous transvenous femoropopliteal bypass to the left leg.
- Wound vac therapy and surgical debridement were performed.

Stage 2: Two weeks later with TOT five times weekly for 1.5 hours/day.
- Complete healing of the right foot by month two
- Near-complete healing of the left foot by month four.

Stage 3: Right SFA stenting and angioplasty.
- TOT was completed,
- By six months, both ulcerations had fully resolved.
- The patient has remained under ongoing multidisciplinary follow-up, with no recurrence at one-year review.

Conclusion

This case illustrates the synergistic benefit of combining endovascular bypass with topical oxygen therapy in complex limb salvage. Through serial interventions and longitudinal management, the patient achieved complete bilateral wound healing within six months and sustained closure at one year, highlighting the potential of integrated perfusion and oxygen strategies in managing advanced vascular and diabetic foot ulcers

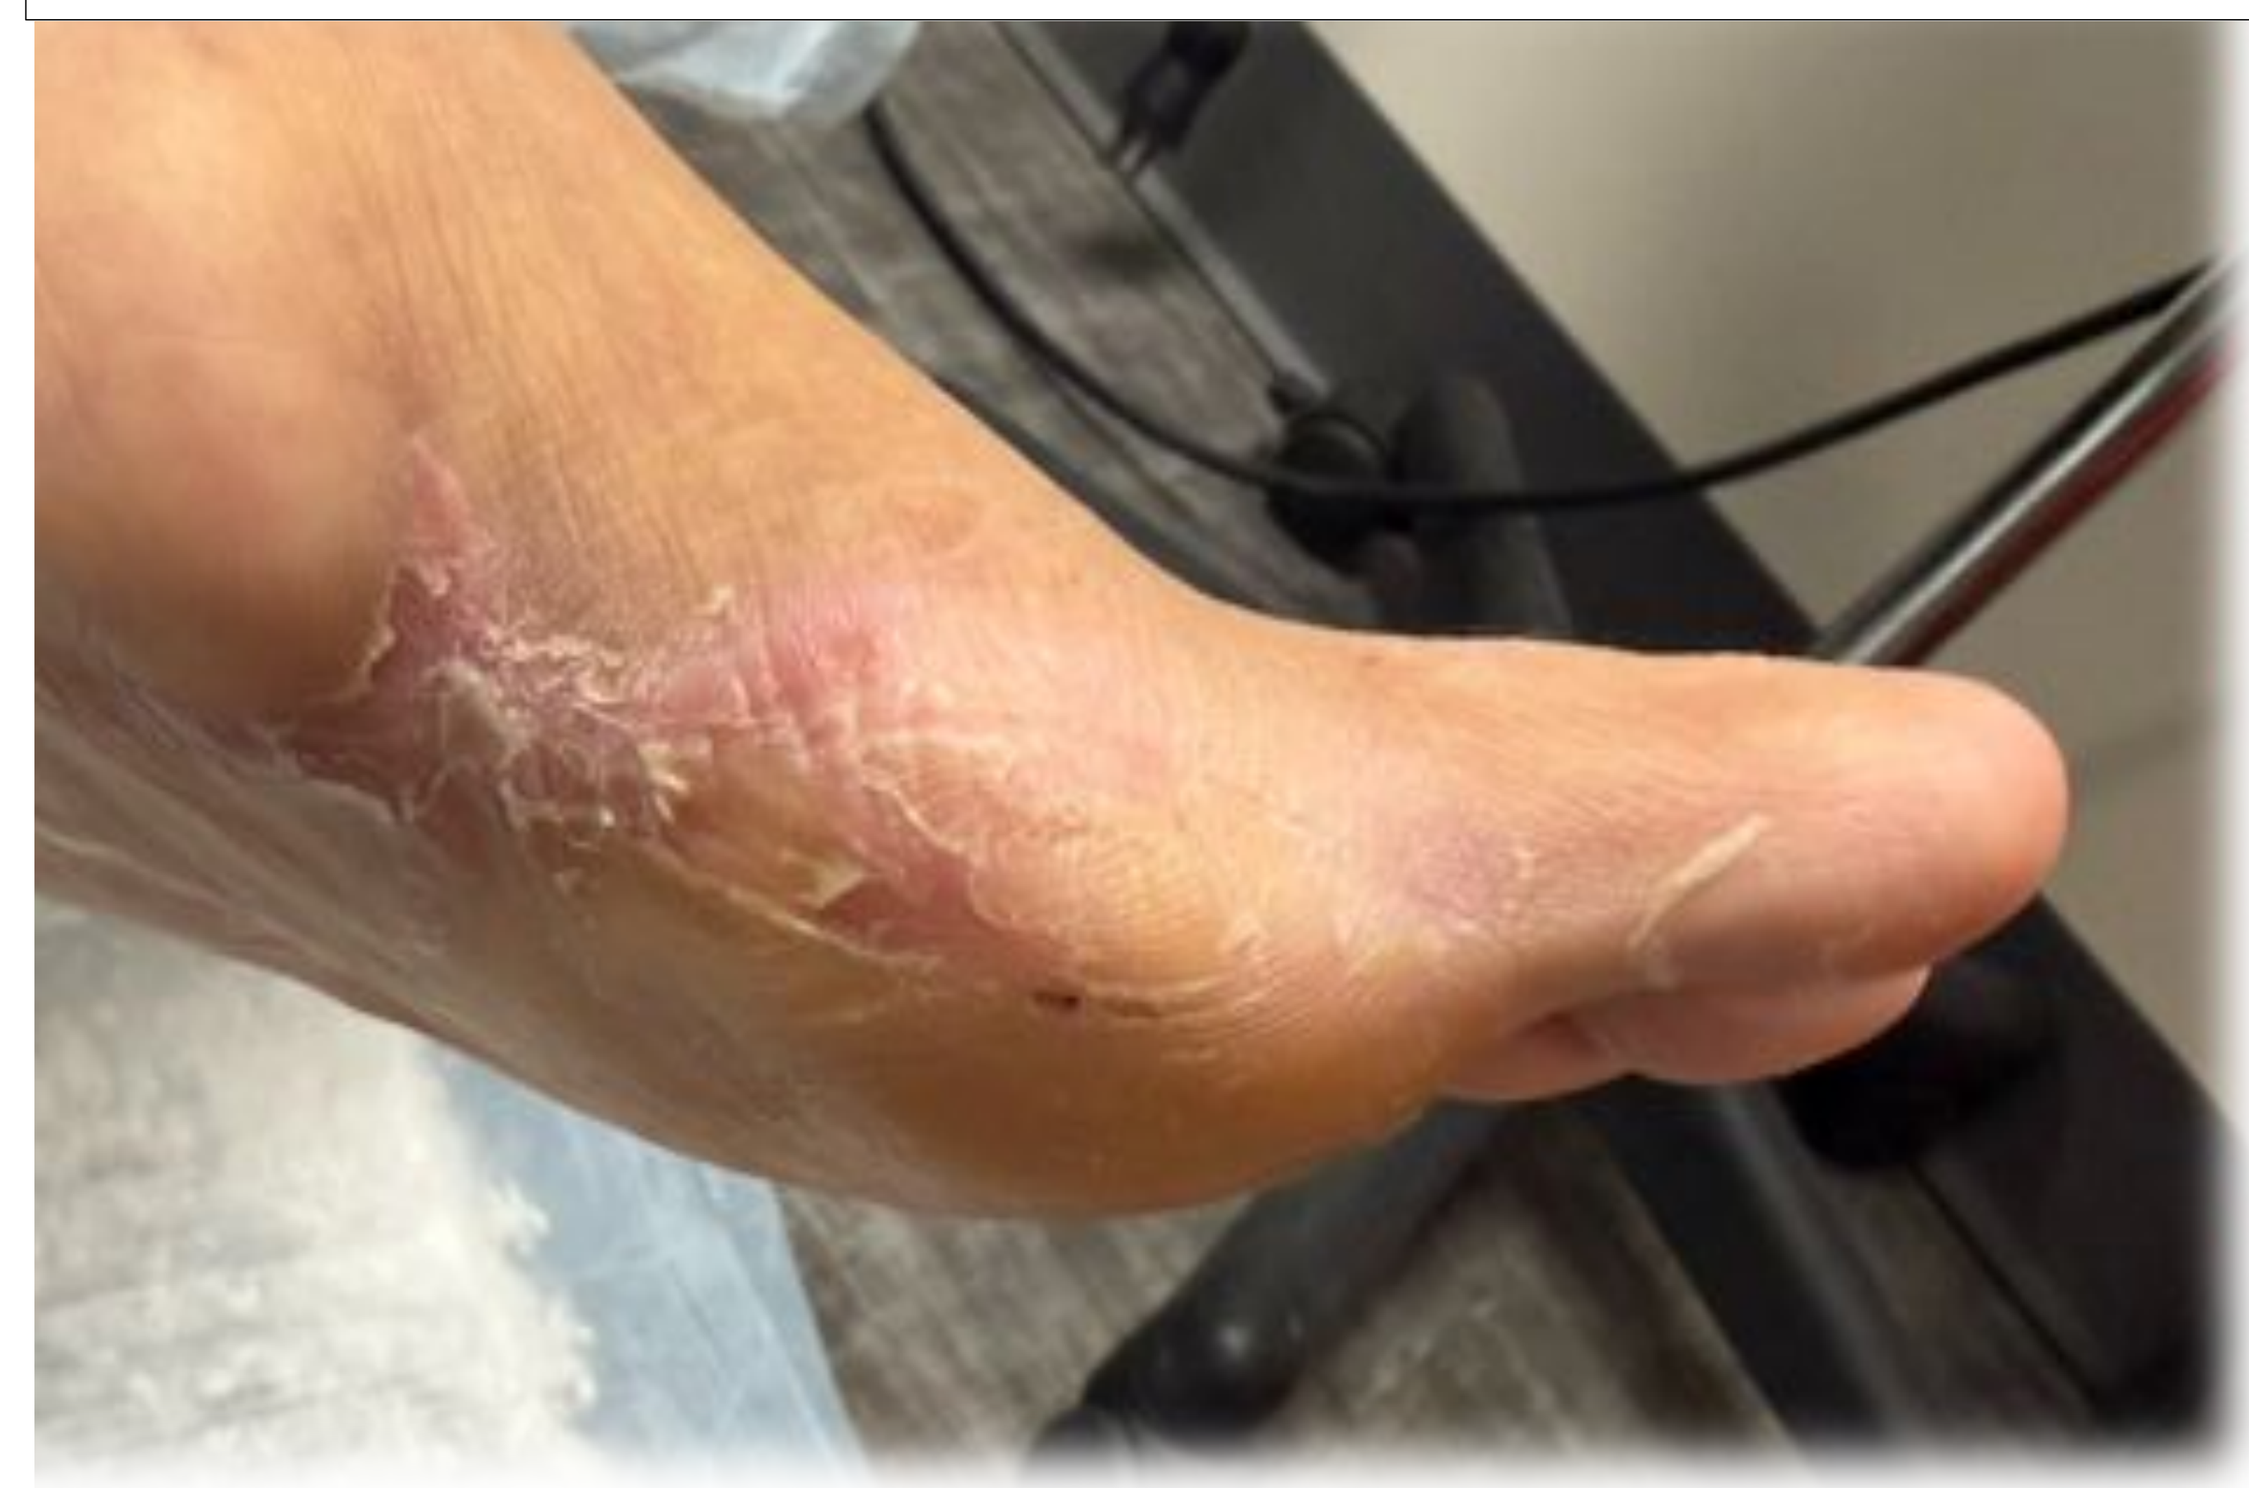

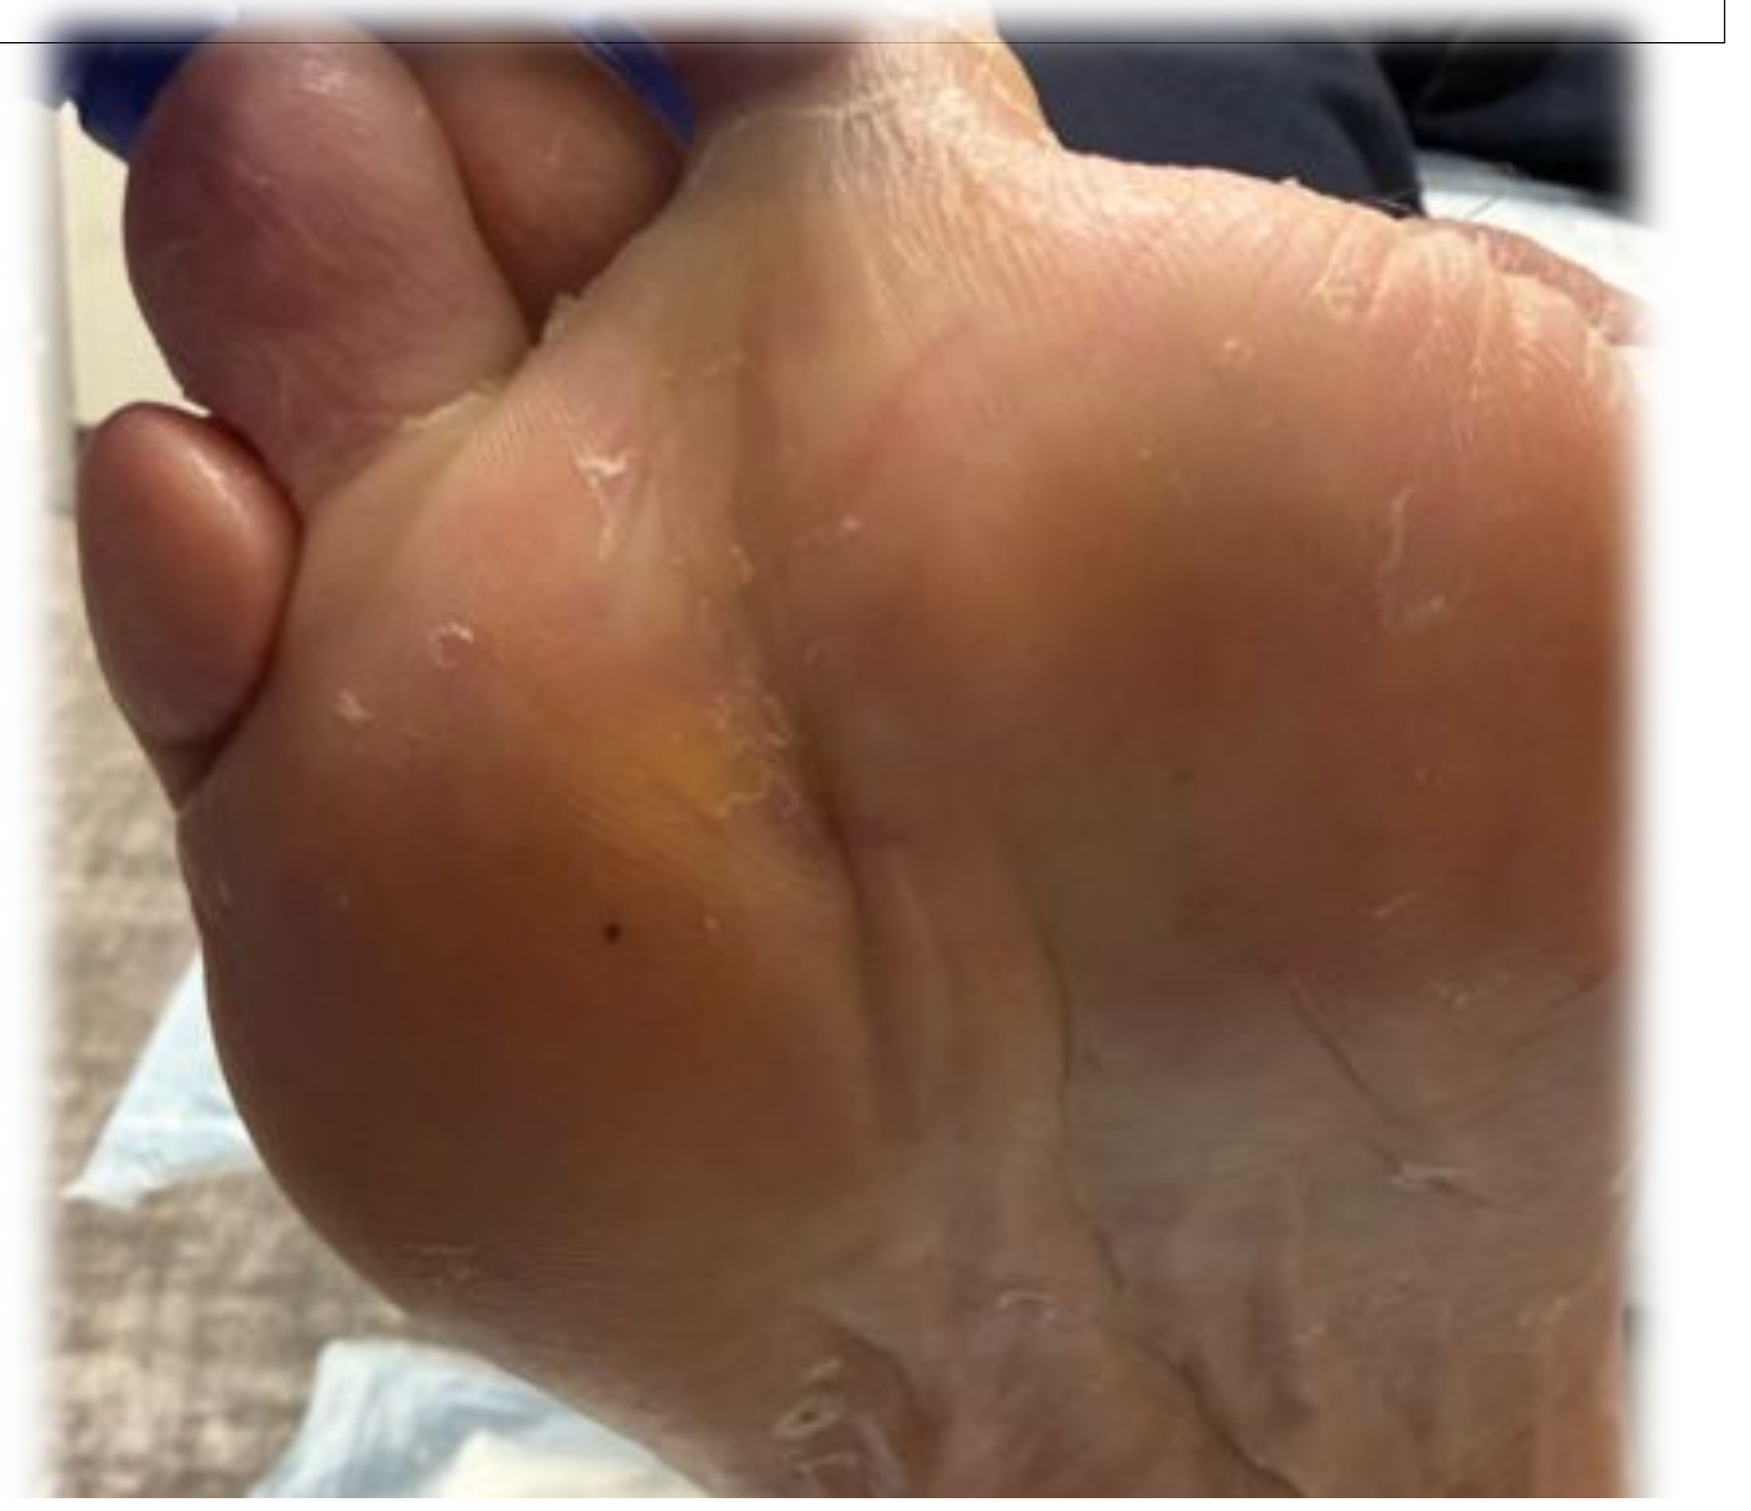

References

1. Dissemond J, Kröger K, Storck M, et al. Topical oxygen wound therapies: An international consensus. J Wound Care. 2023;32(Sup7):S1–S36. doi:10.12968/jowc.2023.32.Sup7.S1
2. Howard MA, Aslam R, Stojadinovic A, et al. Topical oxygen therapy in wound healing: A narrative review of mechanisms and modalities. Auctores Online. 2023. Accessed September 2025. https://auctoresonline.org/article/topical-oxygen-therapy-in-woundhealing-a-narrative-review-of-mechanisms-and-modalities
3. Hayes P, et al. Meta-analysis of topical oxygen therapy for chronic wounds. Int Wound J. 2024;21(3):456–468. doi:10.1111/iwj.14960
4. Cleveland Clinic. Pooled 2-Year Outcomes of DETOUR Trials Support Percutaneous Transfemoral Arterial Bypass. Consult QD. Published May 2024. Accessed September 2025. https://consultqd.clevelandclinic.org/pooled-2-year-outcomes-of-detour-trialssupport-percutaneous-transfemoral-arterial-bypass
5. Endologix Inc. DETOUR2 Trial Final Results Evaluate Endologix DETOUR for PTAB to Treat Complex Femoropopliteal Disease. Endovascular Today. Published March 2025. Accessed September 2025. https://evtoday.com/news/detour2-final-results-evaluateendologix-detour-for-ptab-to-treat-complex-femoropopliteal-diseas